LB-266

Generation of antagonistic anti-TIM-3 and anti-LAG-3 monoclonal antibodies for potential novel immunotherapy combinations

H. Toni Jun, Patricia A. McNeeley, Andrew Lassen, Brandon Hynes, Larry Altobell III, Mark Chhoa, Jesus Olvera, Josh MacLaren, Aldona Balciunas, Minjee Do, Michael Brown, Eric Hare, Audrey D. McConnell, Jean Da Silva Correia, Marilyn Kehry, David King

AnaptysBio, 10421 Pacific Center Court, San Diego, CA 92121

Abstract

Among the most promising approaches in the treatment of cancer is the activation of antitumor immunity by blockade of immune checkpoints. These inhibitory immune checkpoints are crucial for maintaining self-tolerance in the normal immune system but can be co-opted in cancer to allow tumor escape from immune surveillance. Therapeutic validation has been provided using antibodies that inhibit the CTLA-4 and PD-1 signaling pathways, which have shown significant clinical activity. Interestingly, blockade of other T-cell inhibitory signaling molecules TIM-3 and LAG-3 has also been shown to be effective in mouse models of cancer. Potential therapeutic molecules that inhibit the negative signaling of TIM-3 or LAG-3 were identified by screening the AnaptysBio Evolvable Library (ABEL) of fully human germline antibodies. The initial ABEL screens used the soluble extra cellular domains (ECD) of either TIM-3 or LAG-3, followed by screening on cell-surface expressed antigens. The resulting panels of human antibodies were matured to high affinity and potency using SHM-XEL™ which uses mammalian cell display of human IgG followed by in vitro somatic hypermutation (SHM). We have explored the activity of these antibodies both as single agents as well as simultaneous blockade of multiple pathways in in vitro cell-based assays. Inhibition of each pathway in isolation demonstrated immune stimulatory activity as evidenced by increased secretion of IL-2 in a mixed lymphocyte reaction or an activated T-cell assay. Combination of an anti LAG-3 or an anti TIM-3 antibody with an anti-PD-1 inhibitory antibody could increase IL-2 secretion over that seen with blockade of a single checkpoint alone. The magnitude of this effect was donor cell dependent. These data are suggestive that combination immunotherapy towards these targets is worthy of clinical evaluation and may lead to increased efficacy.

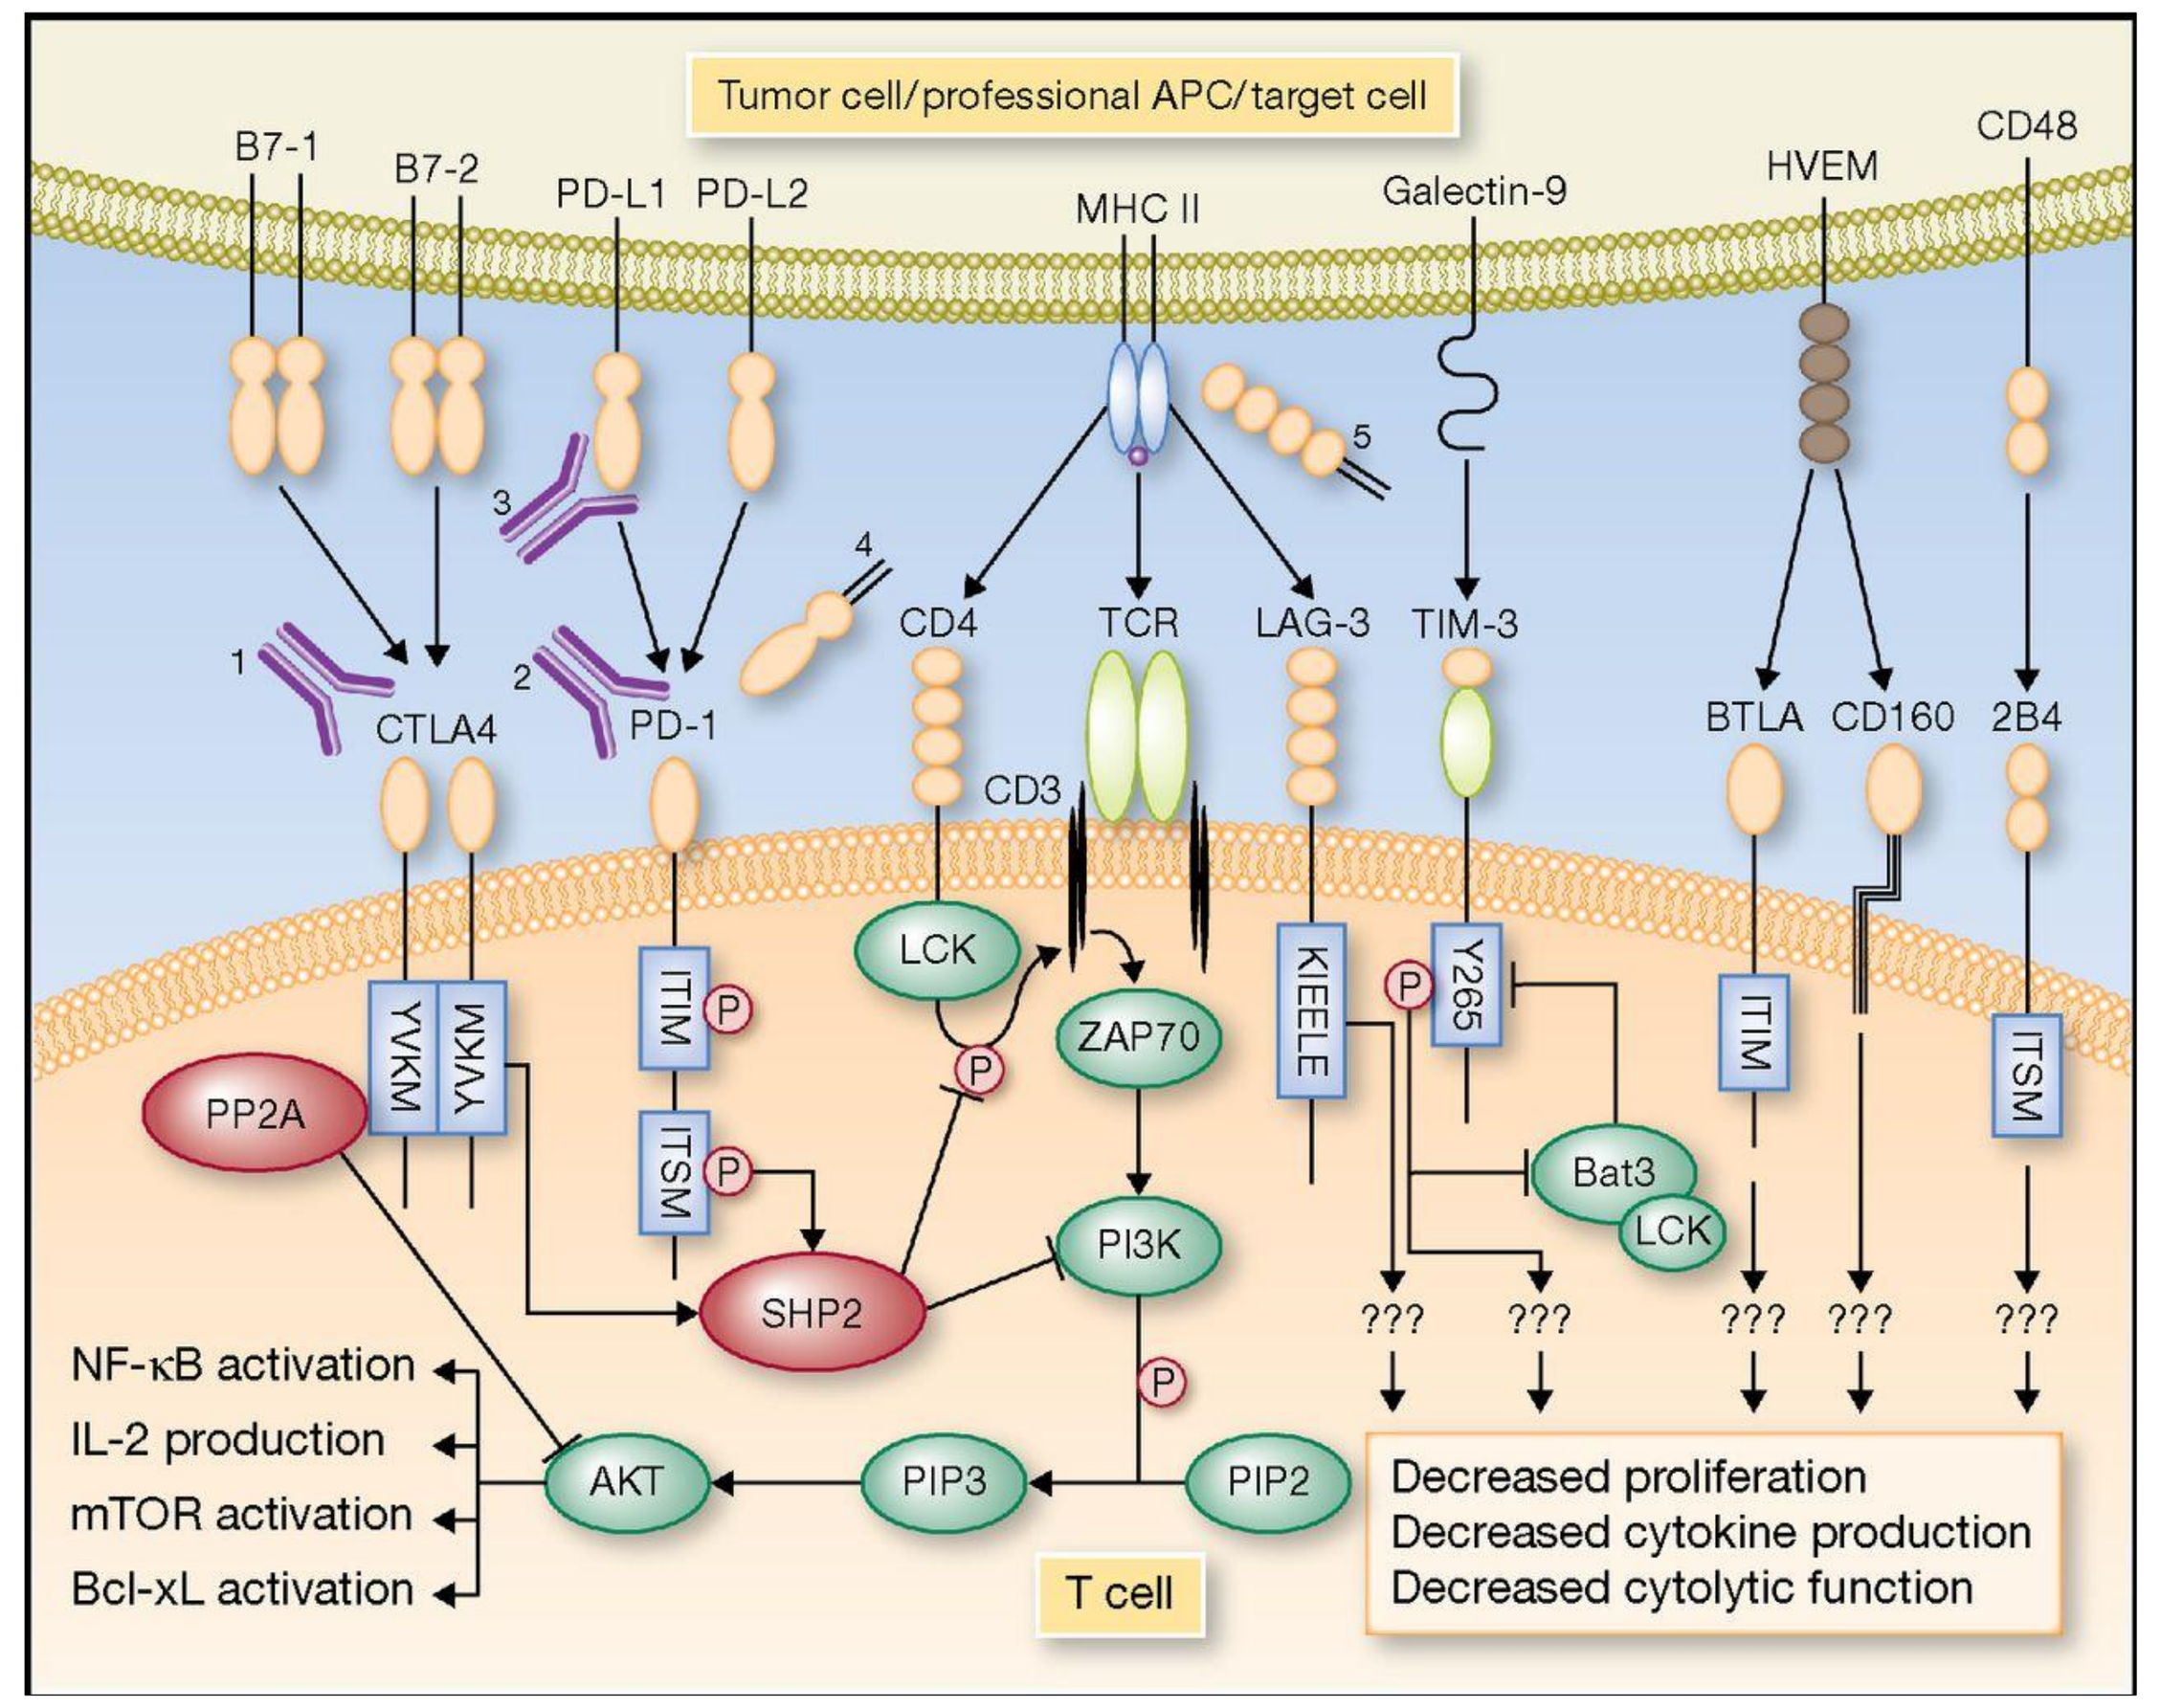
Tumor cell/professional APC/target cell
B7-1
B7-2
PD-L1
PD-L2
MHC II
Galectin-9
HVEM
CD48
CTLA4
PD-1
CD4
TCR
LAG-3
TIM-3
BTLA
CD160
2B4
CD3
LCK
ZAP70
PP2A
YVKM
ITIM
ITSM
KIEELE
Y265
Bat3
SHP2
PI3K
AKT
PIP3
PIP2
NF-κB activation
IL-2 production
mTOR activation
Bcl-xL activation
T cell
Decreased proliferation
Decreased cytokine production
Decreased cytolytic function

Overview of select immune checkpoint molecules.
PD-1 ligation by PD-L1 or PD-L2 results in membrane proximal decreases in TCR signaling. LAG-3 signaling is dependent on interaction with its ligand, MHC II to inhibit T-cell signaling. TIM-3 reportedly binds to Galectin-9, as well as other ligands.

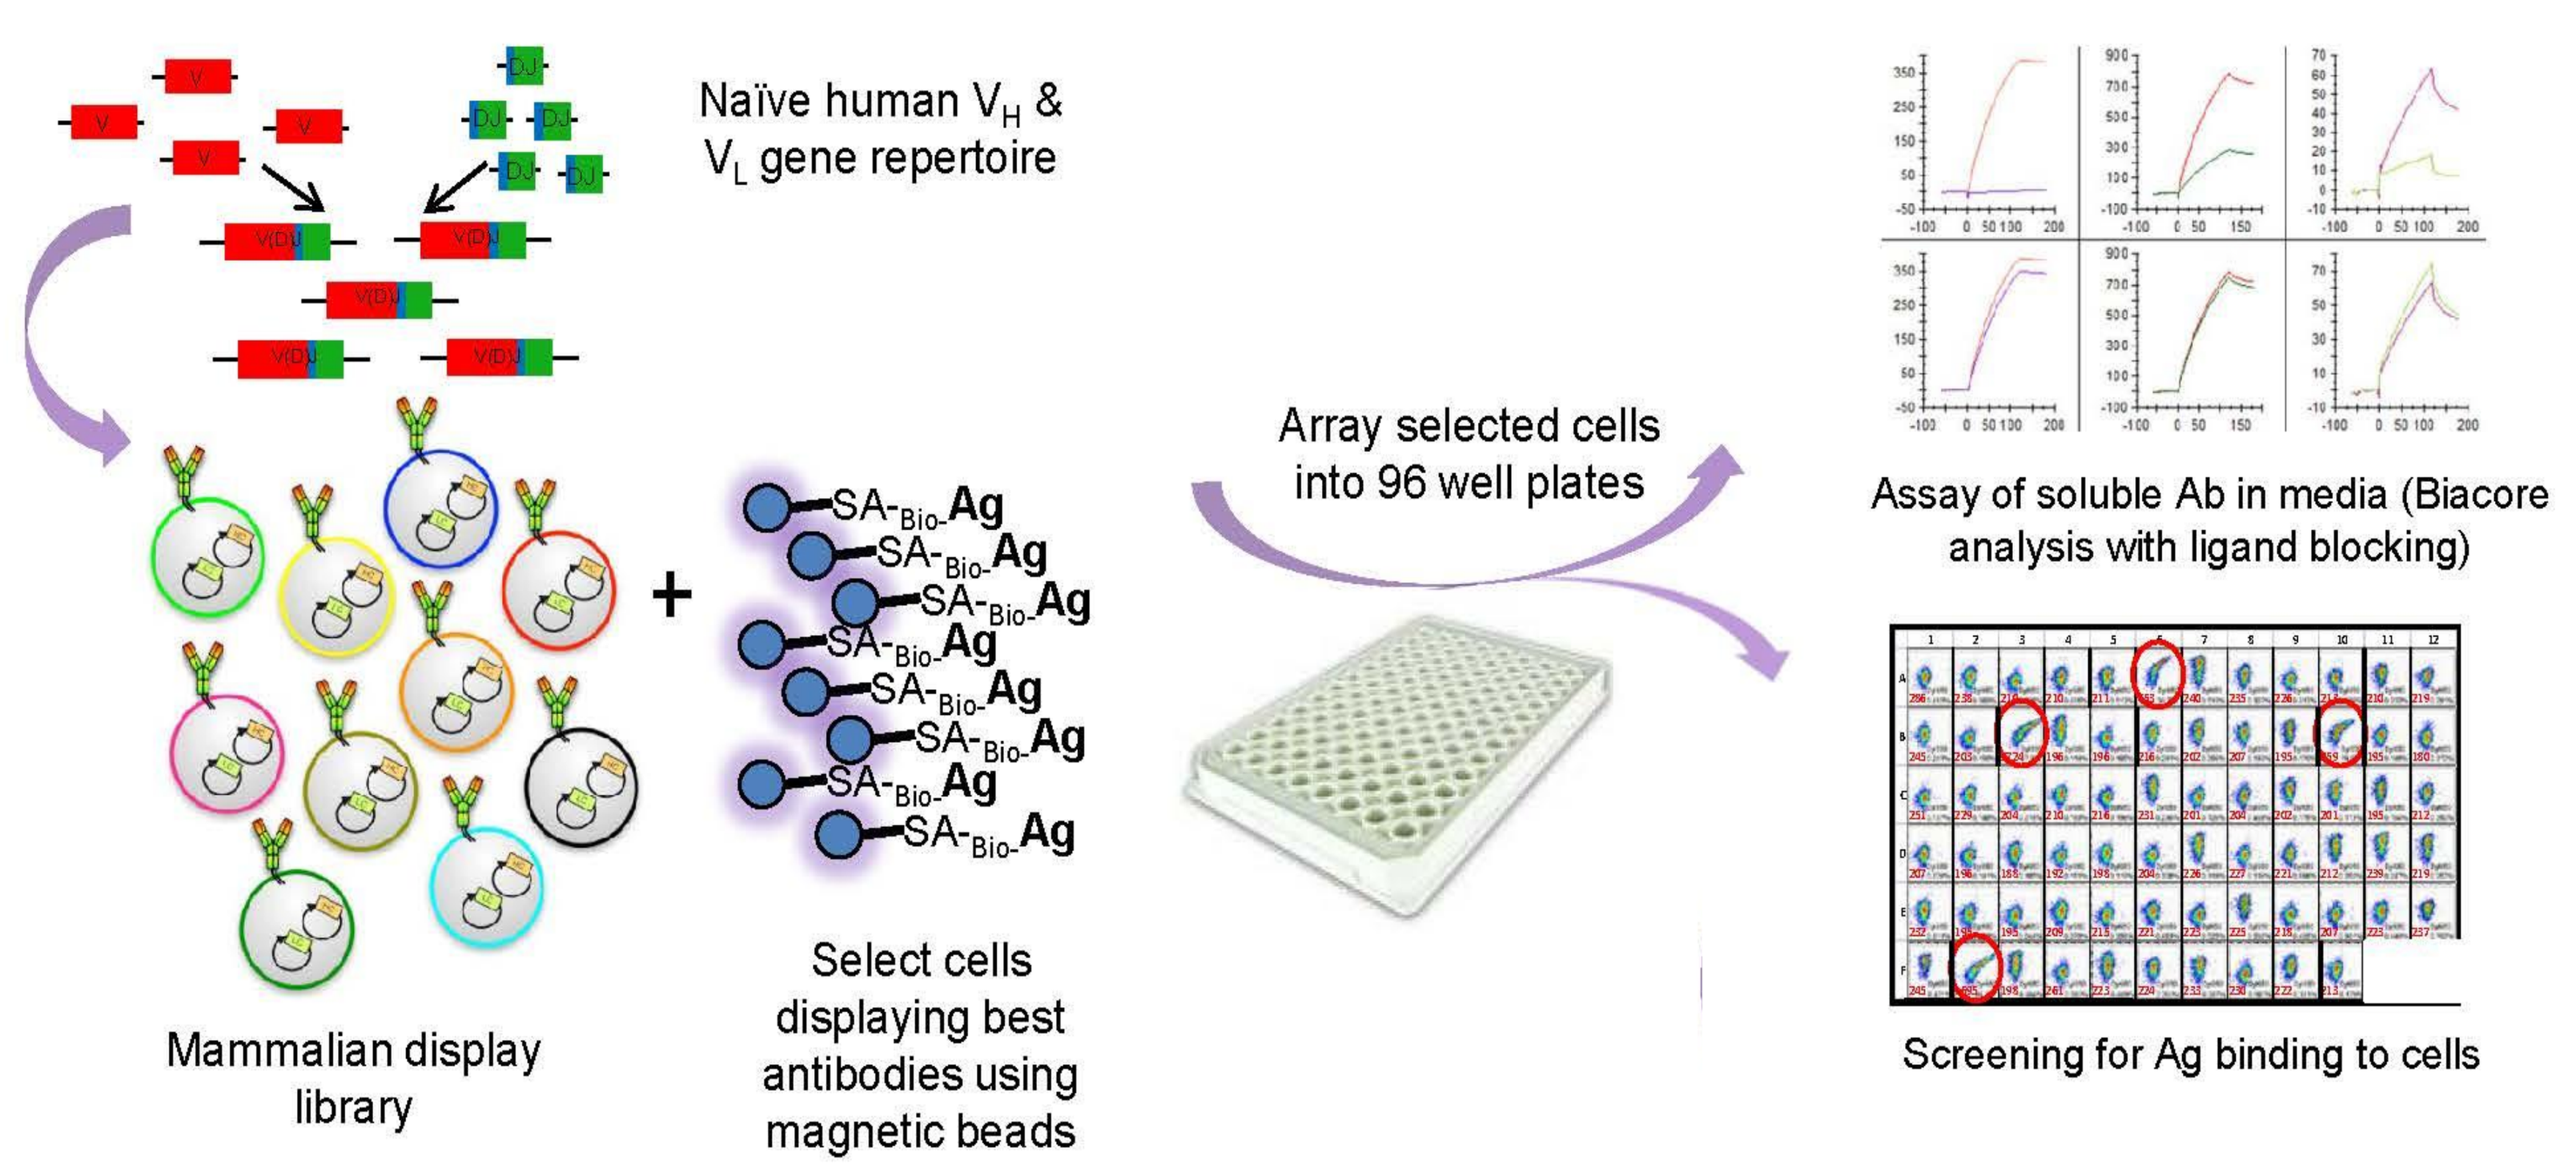
Naïve human V_H & V_L gene repertoire
Mammalian display library
Select cells displaying best antibodies using magnetic beads
Array selected cells into 96 well plates
Assay of soluble Ab in media (Biacore analysis with ligand blocking)
Screening for Ag binding to cells

ABEL library screen overview.
Cells expressing surface-displayed, fully-human antibodies (Abs) that bind to soluble human TIM-3 or LAG-3 ECD were identified from a screening campaign of the ABEL library using magnetic beads coated with huTIM-3 or huLAG-3. A panel of fully-human germline antibodies were isolated that bound specifically to TIM-3 or LAG-3. The heavy (HC) and light (LC) chains of select hits were retransfected to create a stable cell line displaying the mAb on the cell surface. Activation induced deaminase (AID) was then transfected to begin SHM. Iterative rounds of FACS sorting with decreasing concentrations of antigen were performed to identify mAbs with increased affinity for TIM-3 or LAG-3.

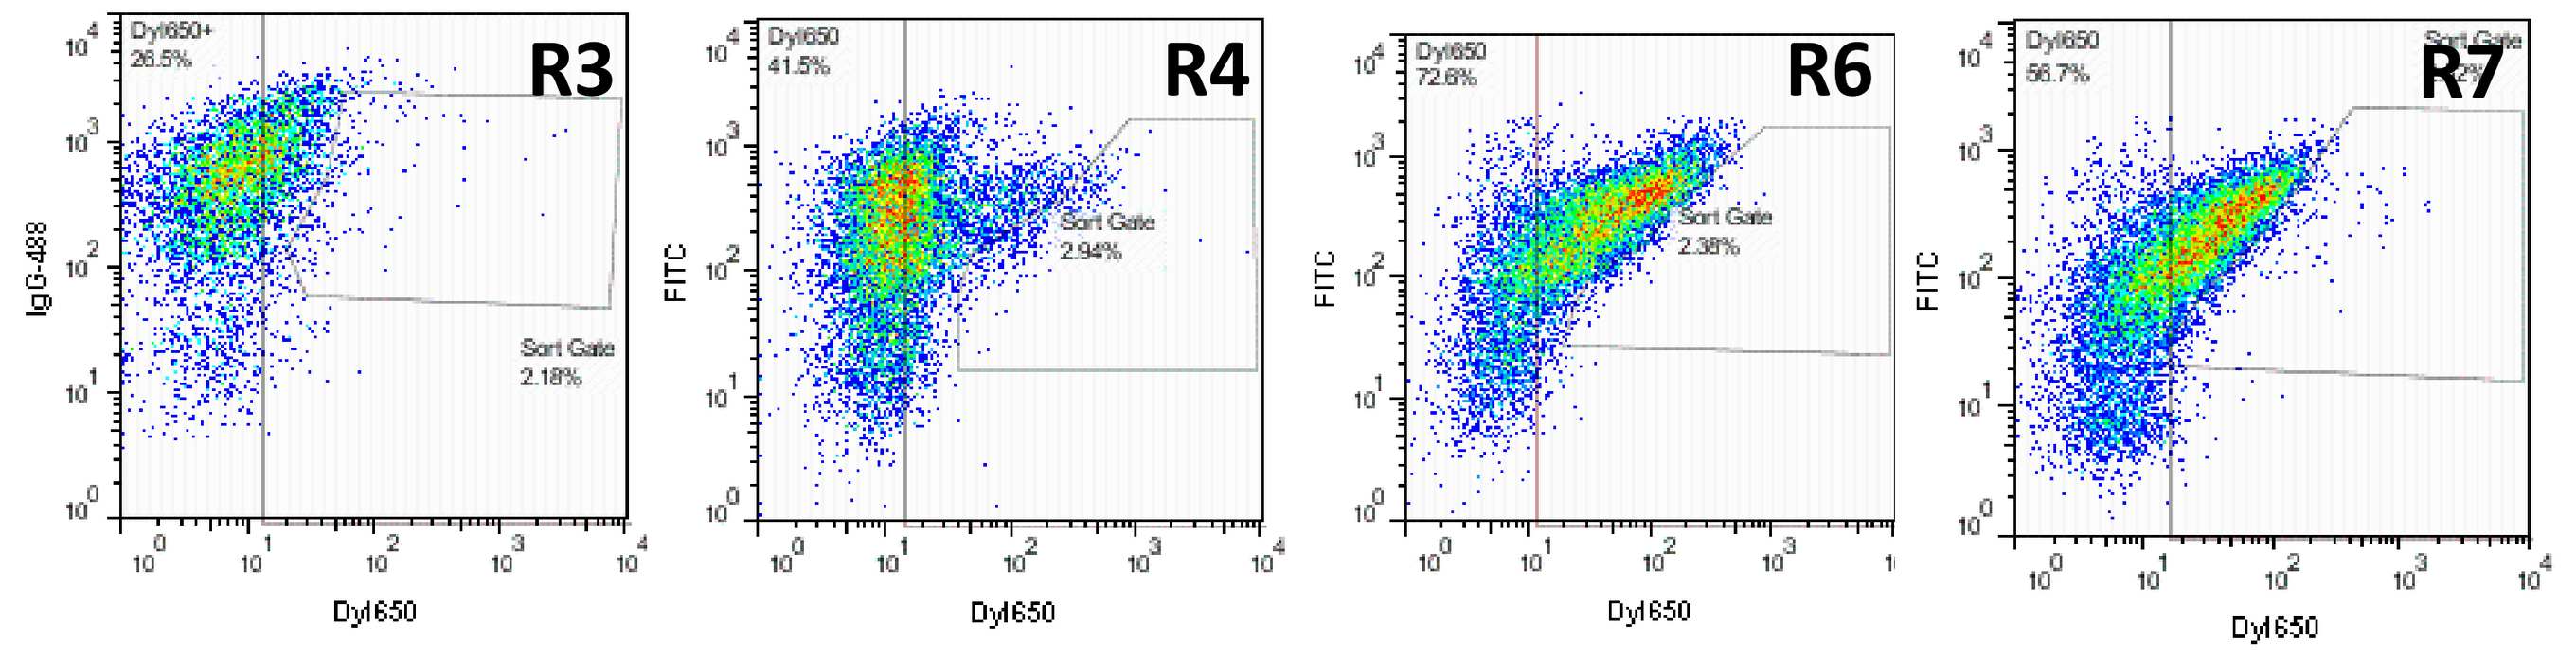
R3
R4
R6
R7
Dyl650
FITC
IgG-488
Sort Gate

Visualization of increased mAb affinity using FACS.
Cells from an evolving population were sorted by flow cytometry following incubation with antigen. The gated population indicated on each FACS plot was expanded and subjected to additional rounds of cell sorting as well as sequence analysis to identify improving mutations.

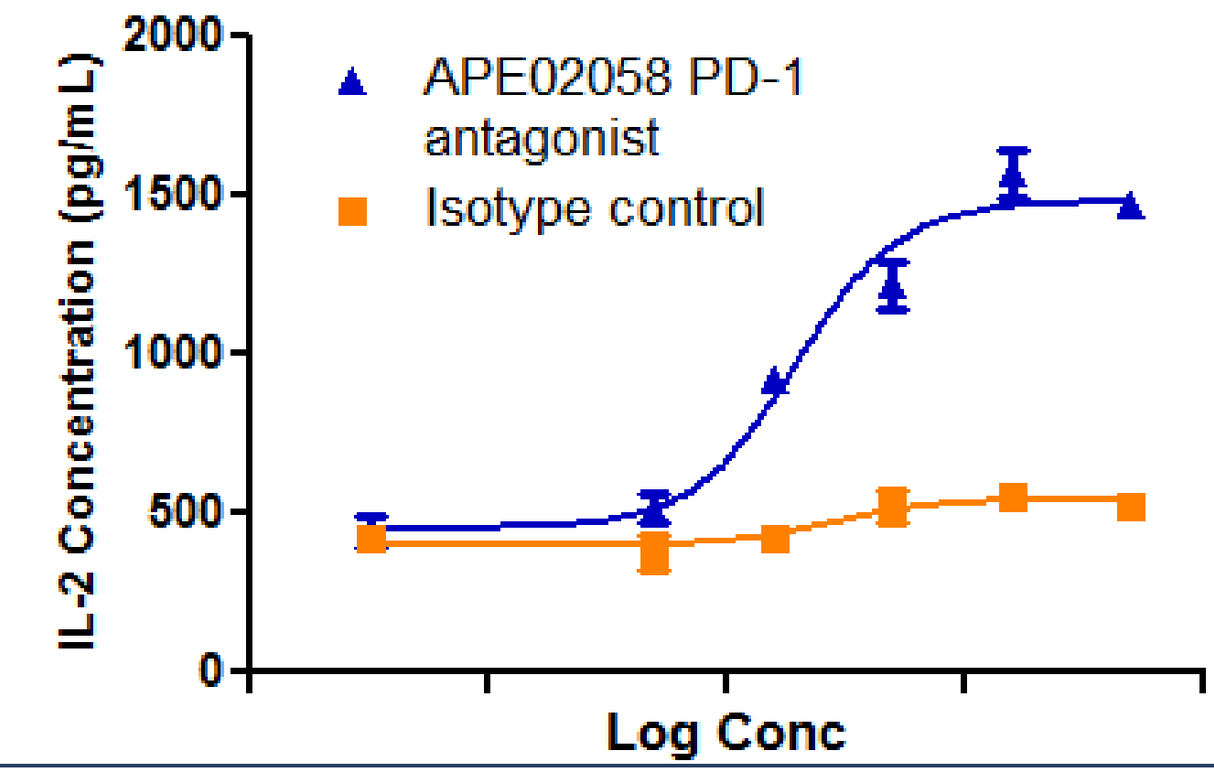
APE02058 PD-1 antagonist
Isotype control
IL-2 Concentration (pg/mL)
Log Conc

Single agent activity of APE02058, an anti-PD-1 antagonist mAb, in a mixed lymphocyte reaction (MLR) assay.
Isolated peripheral blood monocytes from a human donor were differentiated into dendritic cells (DC's) and then mixed with CD4+ T-cells isolated from a second donor. IL-2 levels were measured after 48 hours and graphed above. APE02058, a humanized mAb that displays PD-1 antagonist activity, has an EC50 of **20ng/m**l.

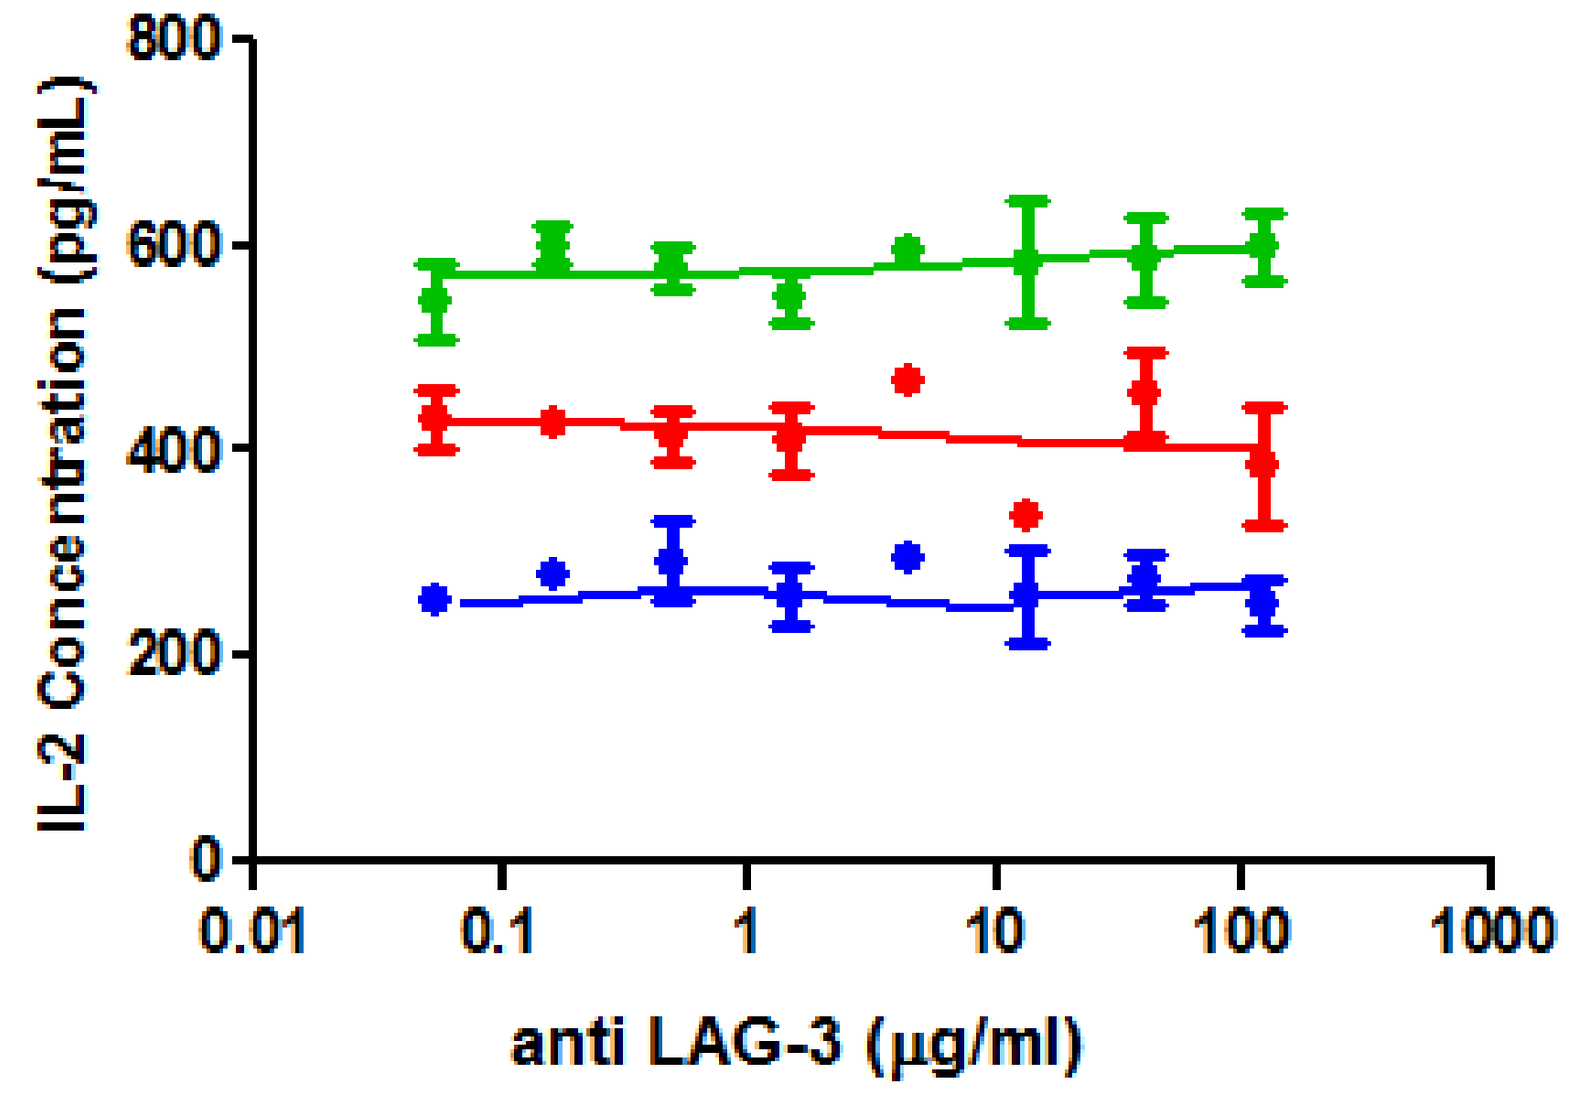
IL-2 Concentration (pg/mL)
anti LAG-3 (μg/ml)

24 hours

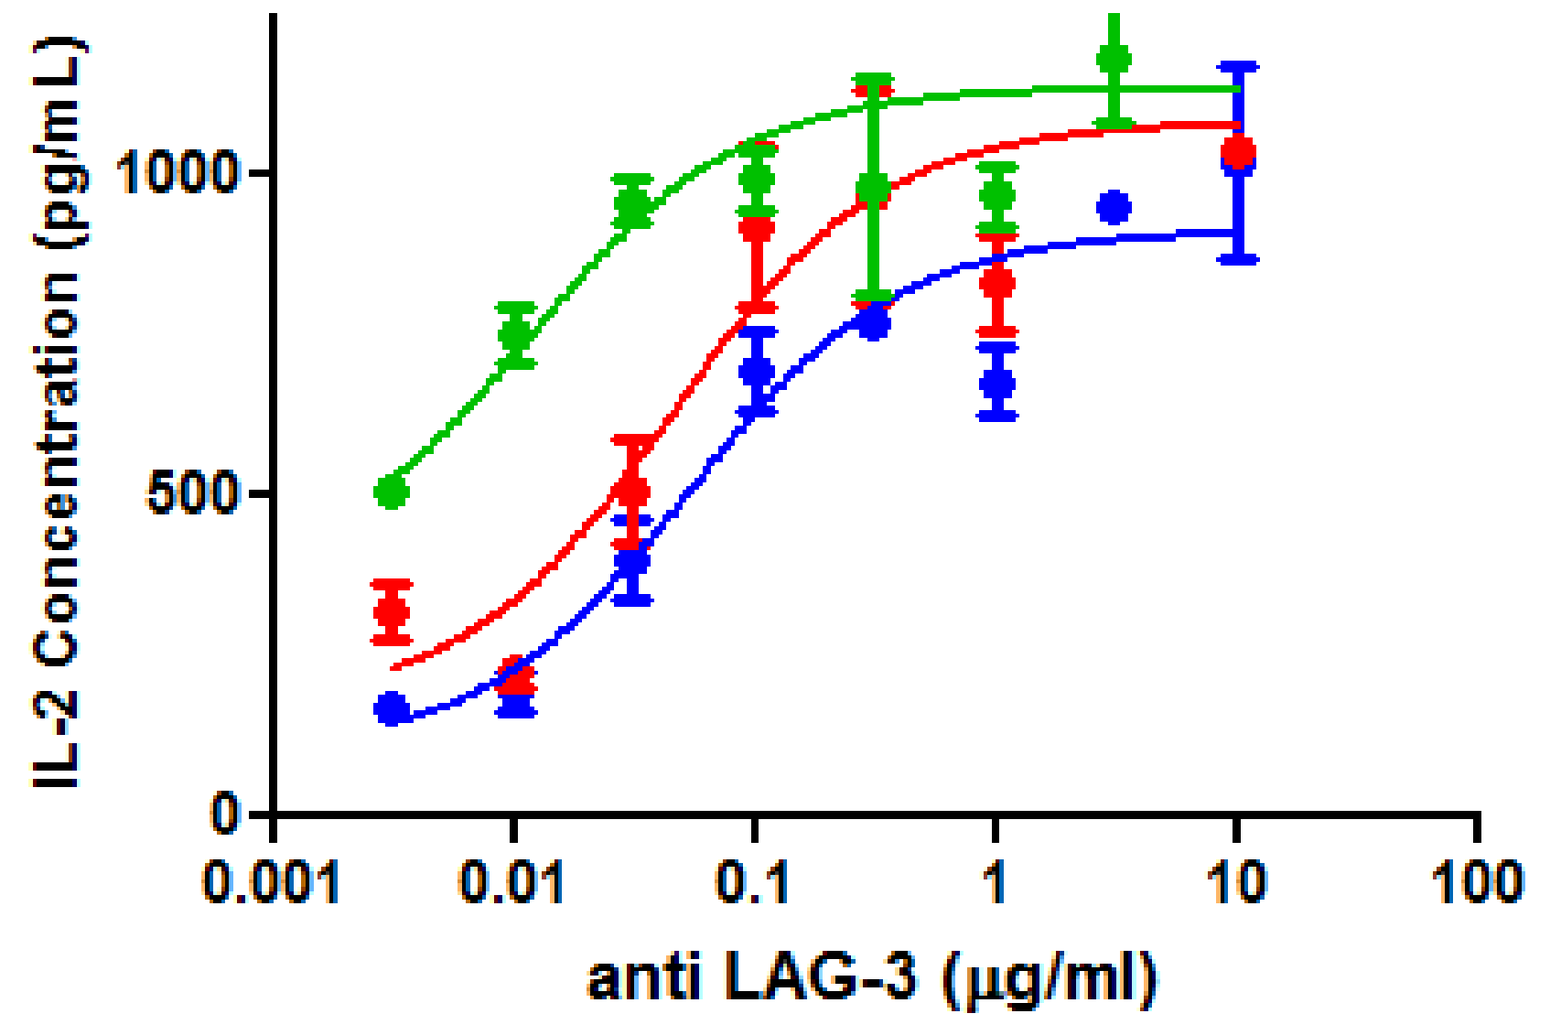
IL-2 Concentration (pg/mL)
anti LAG-3 (μg/ml)

48 hours

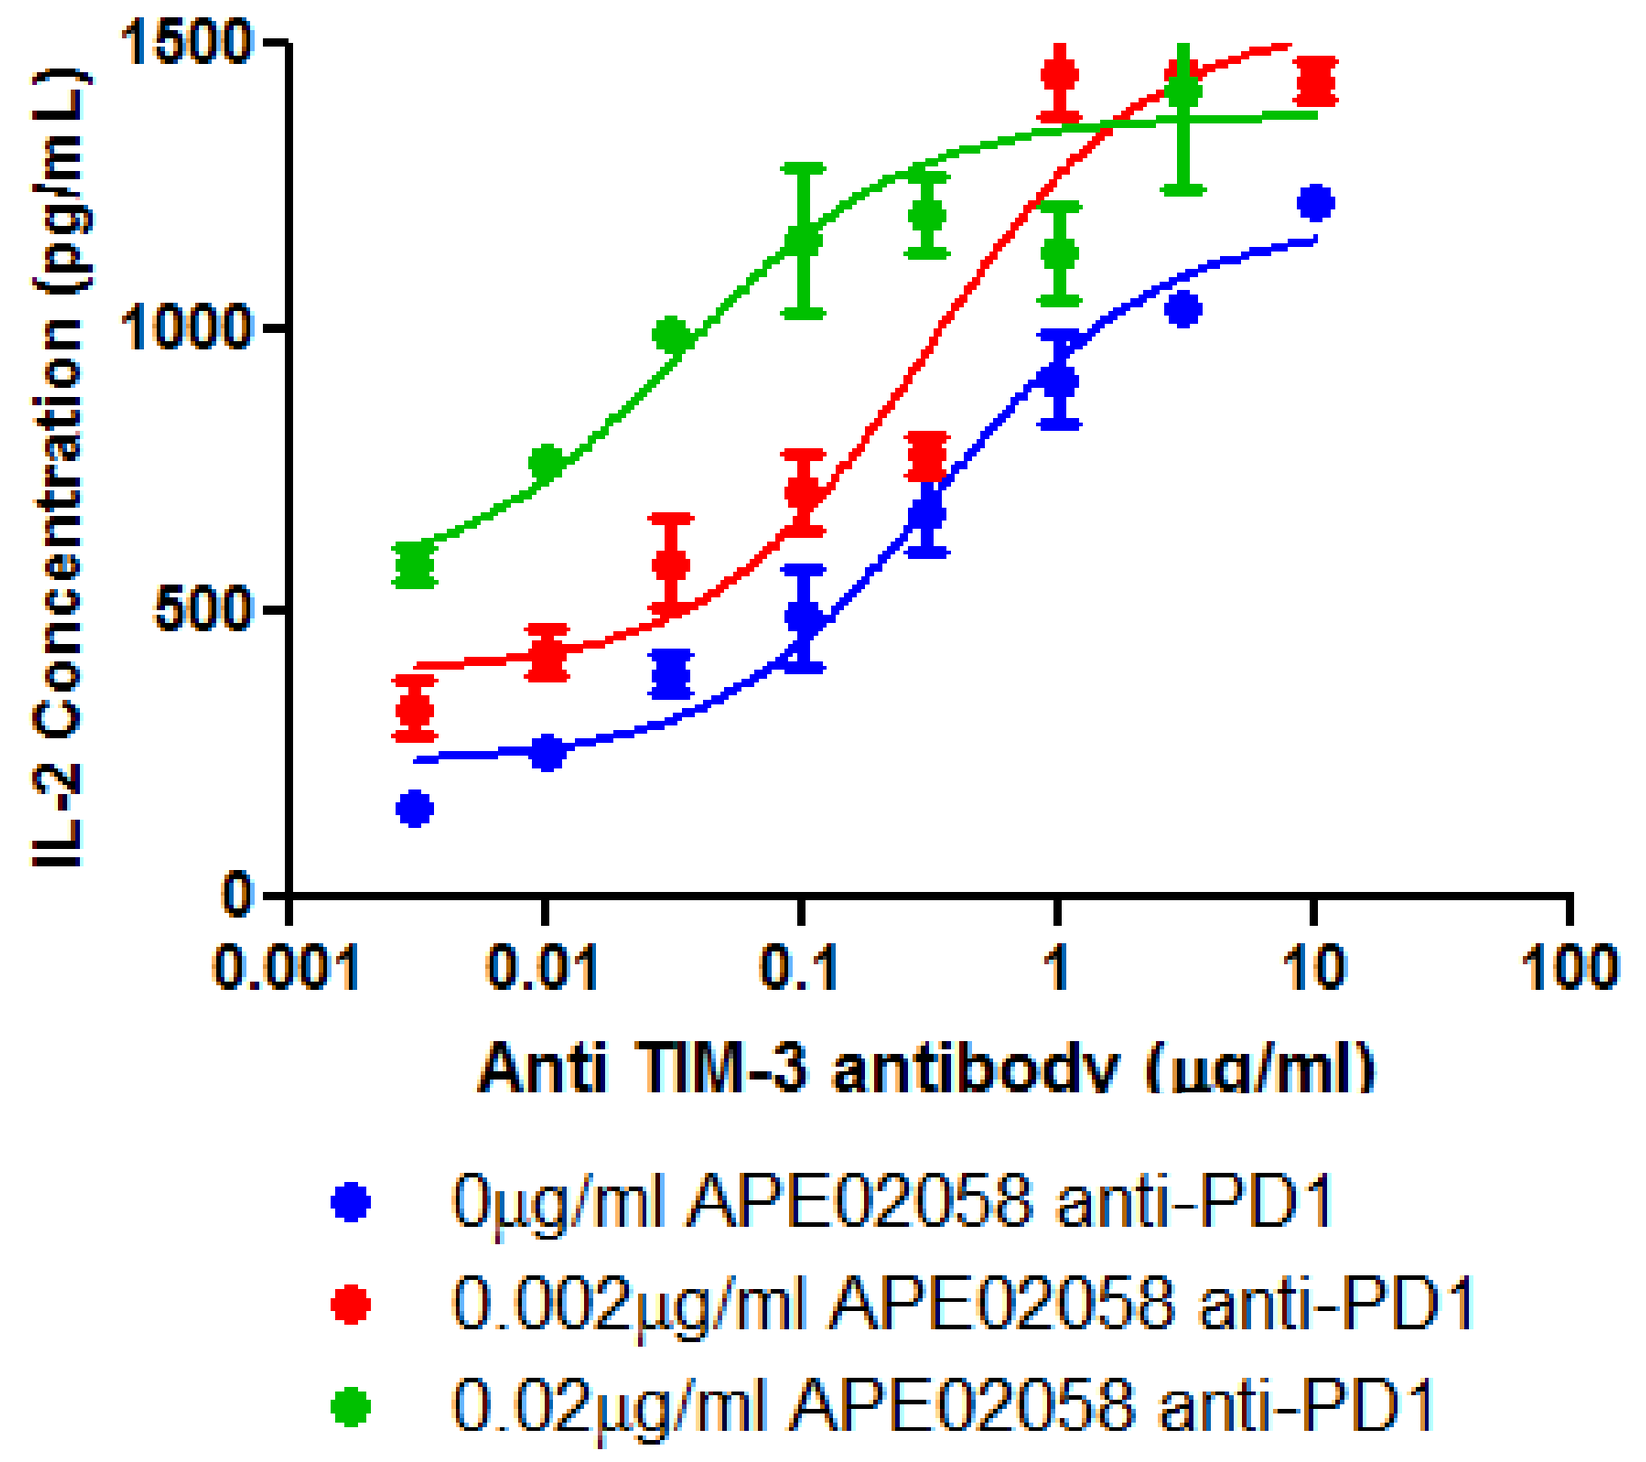
IL-2 Concentration (pg/mL)
Anti TIM-3 antibody (μg/ml)
0μg/ml APE02058 anti-PD1
0.002μg/ml APE02058 anti-PD1
0.02μg/ml APE02058 anti-PD1

48 hours

mAb	Single agent MLR EC50 48 hours	EC50 with 2ng/ml anti-PD-1 (~EC10) 48 hours	EC50 with 20ng/ml anti PD-1 (~EC50) 48 hours
Anti LAG-3	53ng/ml	44ng/ml	11ng/ml
Anti TIM-3	330ng/ml	310ng/ml	33ng/ml
Anti PD-1	20ng/ml	n/a	n/a

Anti-LAG-3 and anti-TIM-3 increase IL-2 secretion both alone and in combination with an anti-PD1 antagonist mAb at **48 hours** in the MLR assay.
Anti LAG-3 or anti TIM-3 mAb was characterized in the MLR assay alone (blue) or in combination with 2ng/ml (red line) or 20ng/ml (green line) of anti-PD-1 antagonist mAb. No effect is observed at 24 hours with PD-1 in combination with anti-LAG-3 (top panel). However, at 48 hours, combination with anti PD-1 increases the potency of both anti-LAG-3 (middle panel) and anti-TIM-3 (bottom panel). EC50 data are summarized in the table.

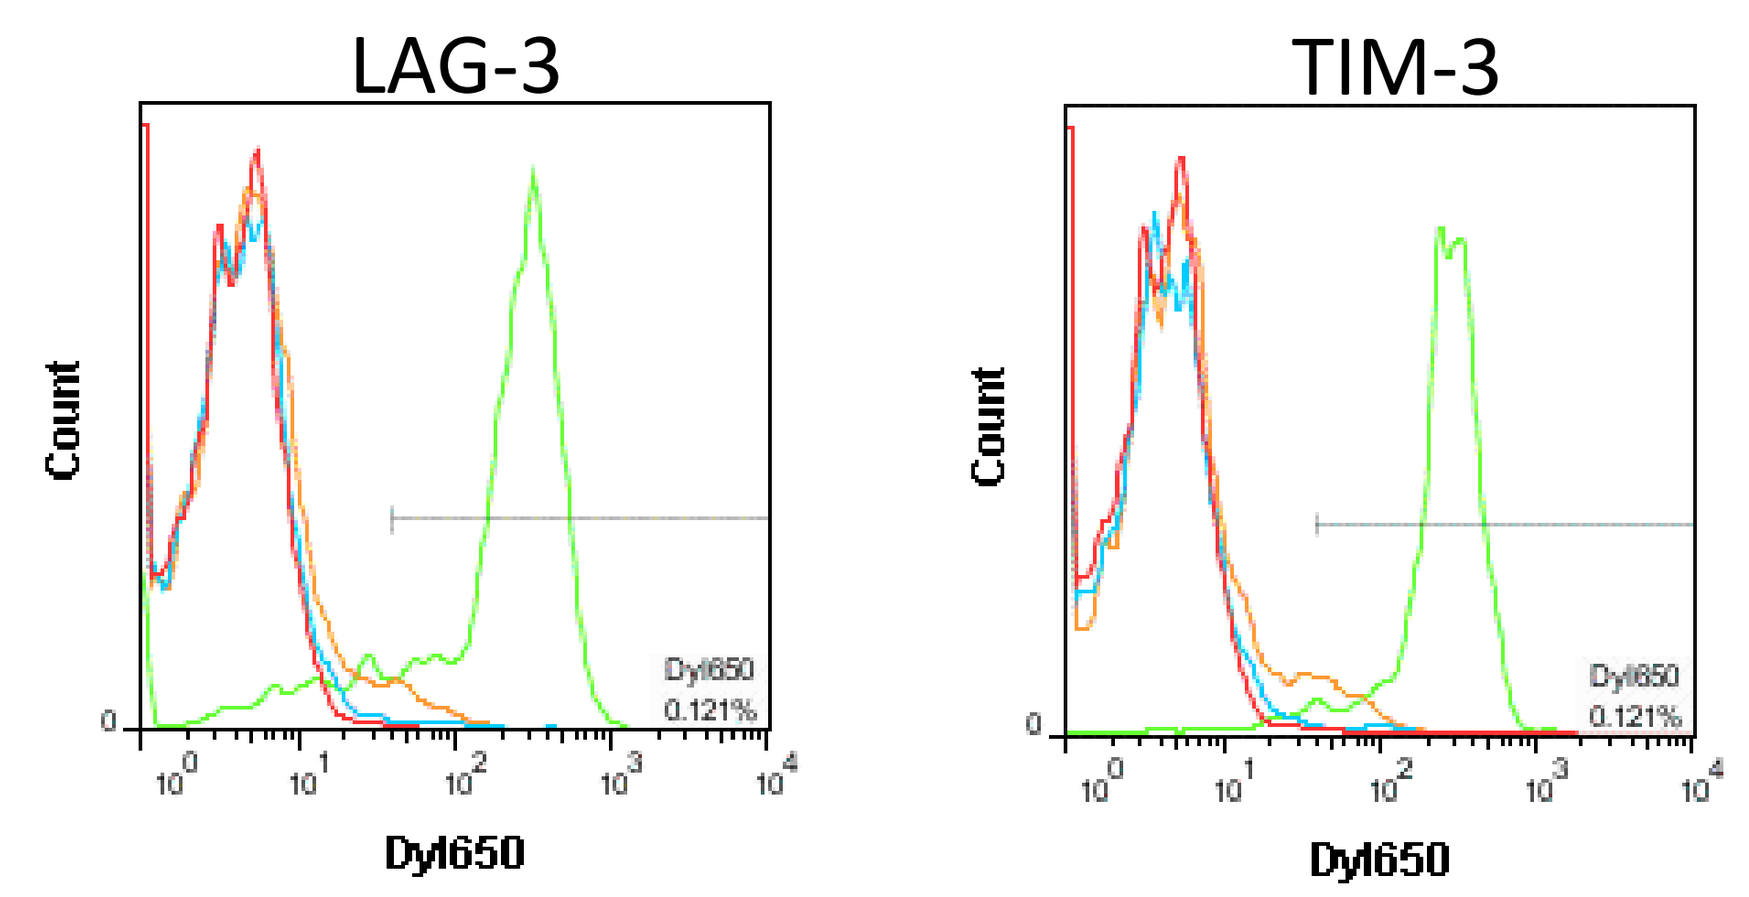
LAG-3
TIM-3
Count
Dyl650

TIM-3 and LAG-3 are both expressed on activated T-cells in the MLR assay.
LAG-3 and TIM-3 expression on T-cells in the MLR assay was examined to assess suitability of the assay for combination studies. $CD4^+$ T-cells (red line) do not express TIM-3 and LAG-3 on the cell surface after purification from human blood. However, 48 hours after culturing with DCs using this donor pair shown above, both TIM-3 and LAG-3 are highly expressed on the T cells (green line), though high donor variability has been observed for TIM-3 and LAG-3 expression using other donors. T cells were also cultured for 48 hours without DCs as a control (blue line). In contrast, PD-1 is expressed on a subset of $CD4^+$ T-cells without DC co-culture and can be upregulated at 24 and 48 hours in the MLR (data not shown), depending upon the donor.

Summary

- Humanization of a mouse mAb combined with in vitro SHM was used to generate an anti-PD-1 antagonist mAb. ABEL library screening combined with in vitro SHM was used to identify functional antagonists to TIM-3.
- In the MLR assay, all three agents demonstrate the ability to increase IL-2 secretion as single agents.
- Anti-TIM-3 or anti-LAG-3 mAb in combination with PD-1 increases IL-2 secretion and decreases the effective EC50 of each single agent.
- These data may suggest dual blockade of immune checkpoints could lead to increased clinical efficacy.

References

1. Bowers, P. M., et al., (2011) Coupling mammalian cell surface display with somatic hypermutation for the discovery and maturation of human antibodies. Proc Natl Acad Sci U S A 108: 20455-20460.
2. Nirschl C J , and Drake C G (2013) Molecular Pathways: Coexpression of Immune Checkpoint Molecules: Signaling Pathways and Implications for Cancer ImmunotherapyClin Cancer Res 19:4917-4924.