50585

ANB032, an Investigational B and T Cell Lymphocyte Attenuator (BTLA) Checkpoint Receptor Agonist, Modulates Dendritic Cell (DC) Maturation and Function: A Novel Mechanism Addressing Atopic Dermatitis Pathophysiology

German Muench, Eric Hare, Pejman Soroosh, Stephen Parmley, Paul Lizzul, Martin Dahl
AnaptysBio, Inc., San Diego, CA, USA

ABSTRACT

Atopic Dermatitis (AD) is characterized by heterogeneous immunologic drivers including broad T cell (Th1, Th2, Th17, Th22) and DC activation. BTLA is a co-inhibitory checkpoint receptor that regulates T cell, B cell and DC function. ANB032, a BTLA agonist antibody, has been shown to inhibit activated T cell proliferation (Th1, Th2, Th17, Th22), and reduce production of inflammatory cytokines in AD patient-derived PBMCs[1]. DCs represent a heterogeneous population of myeloid lineage playing a pivotal role in initiating adaptive immune responses and maintaining immune tolerance. Although the expression of BTLA on subsets of DCs has been reported, BTLA's role in modulating DC maturation and function has not been thoroughly investigated. To address the functional role of BTLA on DCs, an LPS-mediated maturation assay with monocyte-derived DCs was performed to confirm that mature DCs highly express BTLA. ANB032 reduced HLA-DR expression, co-stimulatory molecule expression, and inhibited inflammatory cytokine production from DCs challenged with LPS. When co-cultured with allogenic naïve T cells, ANB032-treated DCs increased the generation of Foxp3+ Tregs and decreased production of Th1 and Th2 cytokines. BTLA agonism by ANB032 inhibits a broad range of immune cells and modulates DC function, while inducing Tregs, and potentially restoring immune balance, which may provide therapeutic value in treating autoimmune and inflammatory diseases. A double-blind, placebo-controlled, global Phase 2 study of ANB032 in moderate-to-severe AD is actively enrolling subjects (NCT05935085).

BACKGROUND AND OBJECTIVE

- AD is a systemic, heterogenous inflammatory disease whose pathogenesis is driven by Th1, Th2, Th17, Th22, and DCs in tissue and periphery
- There are significantly more DCs in the skin of AD patients, with up to 10-fold increase in the epidermis and up to 3.5-fold increase in the dermis[2]
- BTLA is a co-inhibitory checkpoint receptor expressed preferentially on activated T cells, B cells, and DCs, key contributors to inflammatory diseases
- Preclinical studies demonstrated that BTLA-deficient mice show increased T cell proliferation and susceptibility to spontaneous development of autoimmune diseases, including dermatitis, further supporting data that BTLA negatively regulates T cell activation and proliferation[3,4]
- Although the expression of BTLA on subsets of DCs has been reported, BTLA's role in modulating DC maturation and function has not been thoroughly investigated
- ANB032 is an investigational non-depleting BTLA agonist antibody that does not compete with the binding of BTLA to herpesvirus entry mediator (HVEM), its ligand (**Figure 1**)

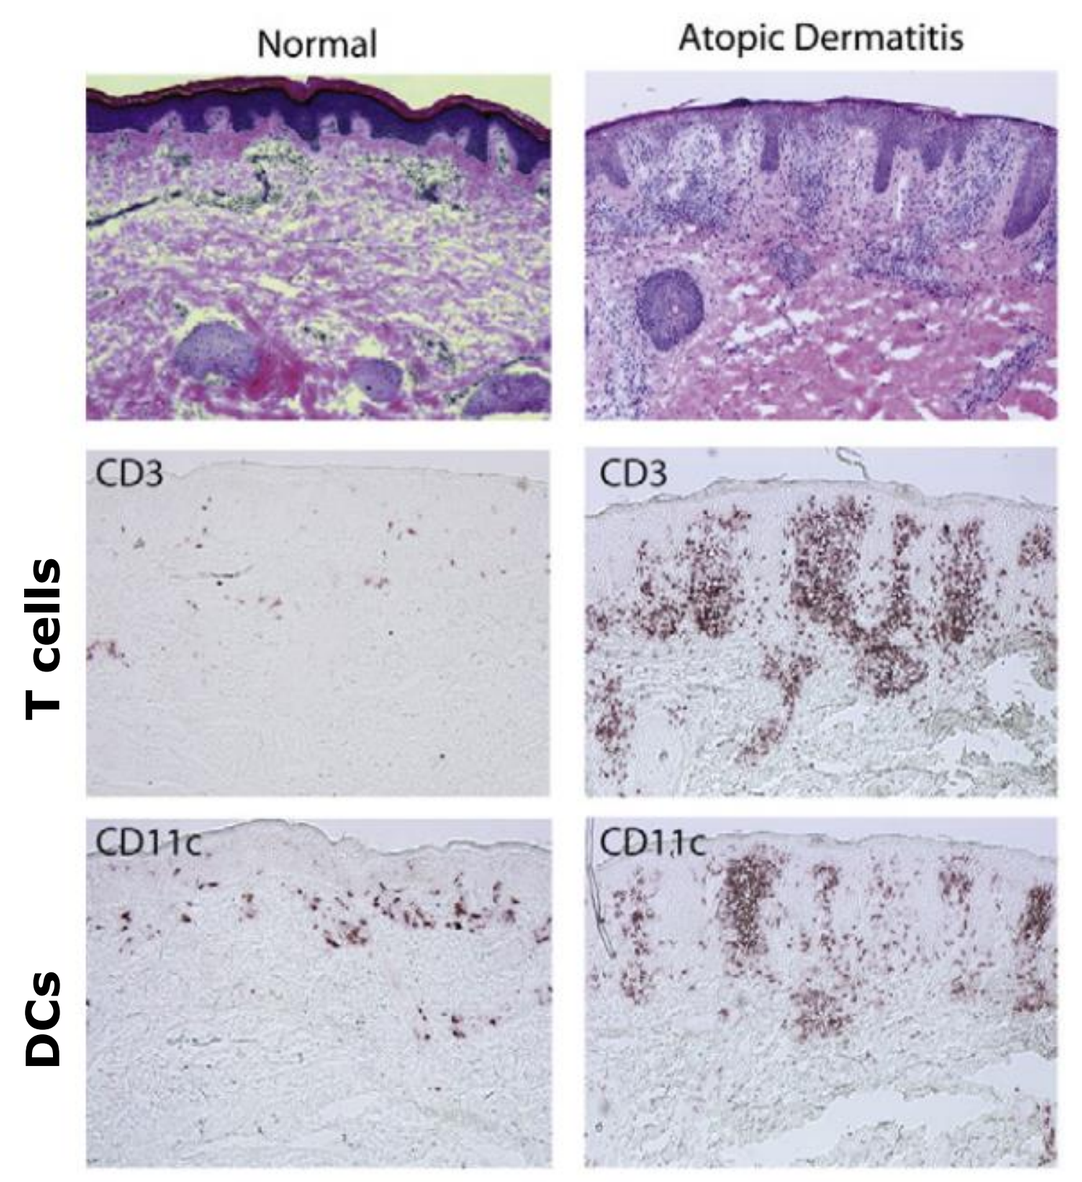
Normal
Atopic Dermatitis
T cells
DCs
CD3
CD3
CD11c
CD11c

Guttman-Yassky et al, JACI, 2011.

Proposed Mechanism of Action for ANB032

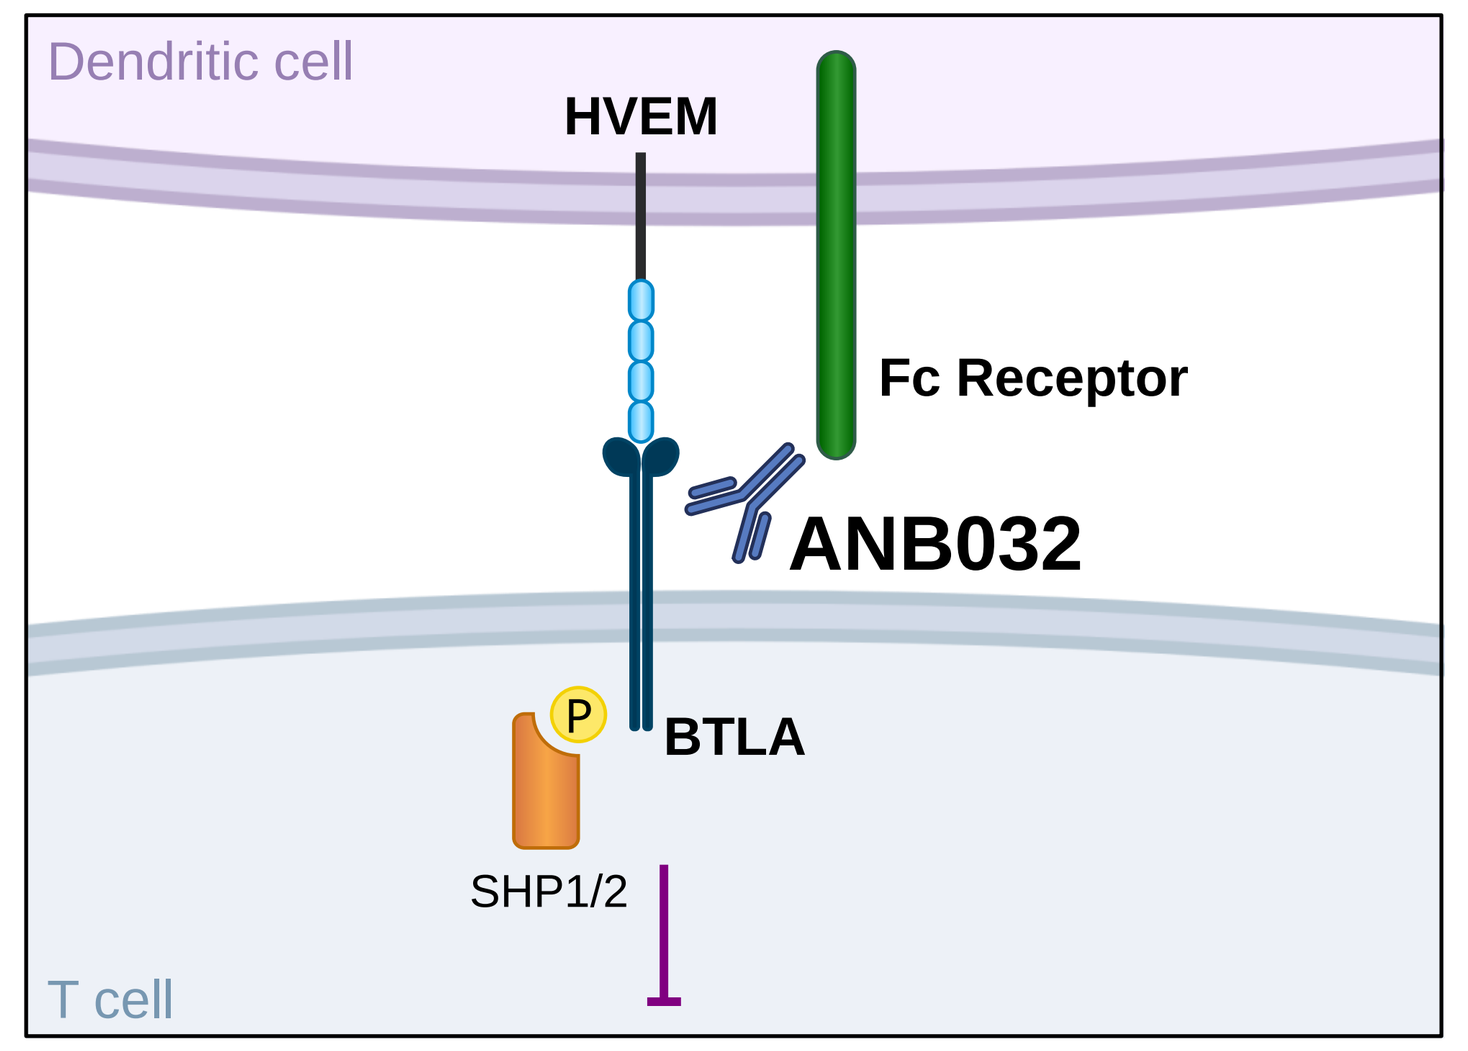
Dendritic cell
HVEM
Fc Receptor
ANB032
P
BTLA
SHP1/2
T cell

Figure 1. Schematic of proposed MOA

BTLA is a key node of immune regulation

- BTLA is a potent checkpoint receptor
- Expressed only on immune cells and preferentially on activated immune cells
- Dysregulation of BTLA pathway accelerates onset and exacerbates disease

ANB032: IgG4 antibody (non-depleting)

- Binds BTLA proximal to immune cell
- Fc receptor binding contributes to differentiated potency
- Non-blocking of HVEM engagement

Objective: Investigate the role of BTLA and effect of ANB032 on DC maturation and activation in a preclinical model

METHODS

<u>BTLA expression on DCs:</u>

- Purified monocytes from healthy PBMCs were differentiated to DCs
- Differentiated DCs were either stimulated with lipopolysaccharide (LPS) or rested in fresh medium, then stained for MHC II and CD11c, and the BTLA expression was evaluated on immature and mature DCs

<u>Effect of ANB032 on DCs:</u>

- Differentiated DCs were treated with either ANB032 or isotype control, then stimulated with LPS and stained for MHC II and CD11c to evaluate the maturation state of DCs, absolute number of mature DCs, expression of MHC II and activation markers

<u>Effect of ANB032 on Tregs and inflammatory cytokines:</u>

- Differentiated DCs were treated with either ANB032 or isotype control
- DCs were co-cultured with allogenic naïve CD4 T cells, then T cells were stained for CD4, CD25, and intracellular Foxp3 to identify inducible regulatory T cells (iTreg)
- The absolute number of differentiated iTregs and secretion of inflammatory cytokines were evaluated by FACS and MSD, respectively

RESULTS

BTLA was Highly Expressed on Mature DCs with LPS Stimulation

- LPS-stimulated DCs resulted in 40% mature and 50% immature DCs
- BTLA expression was seen in 92.7% of mature DCs versus 9.3% of immature DCs (**Figure 2**)

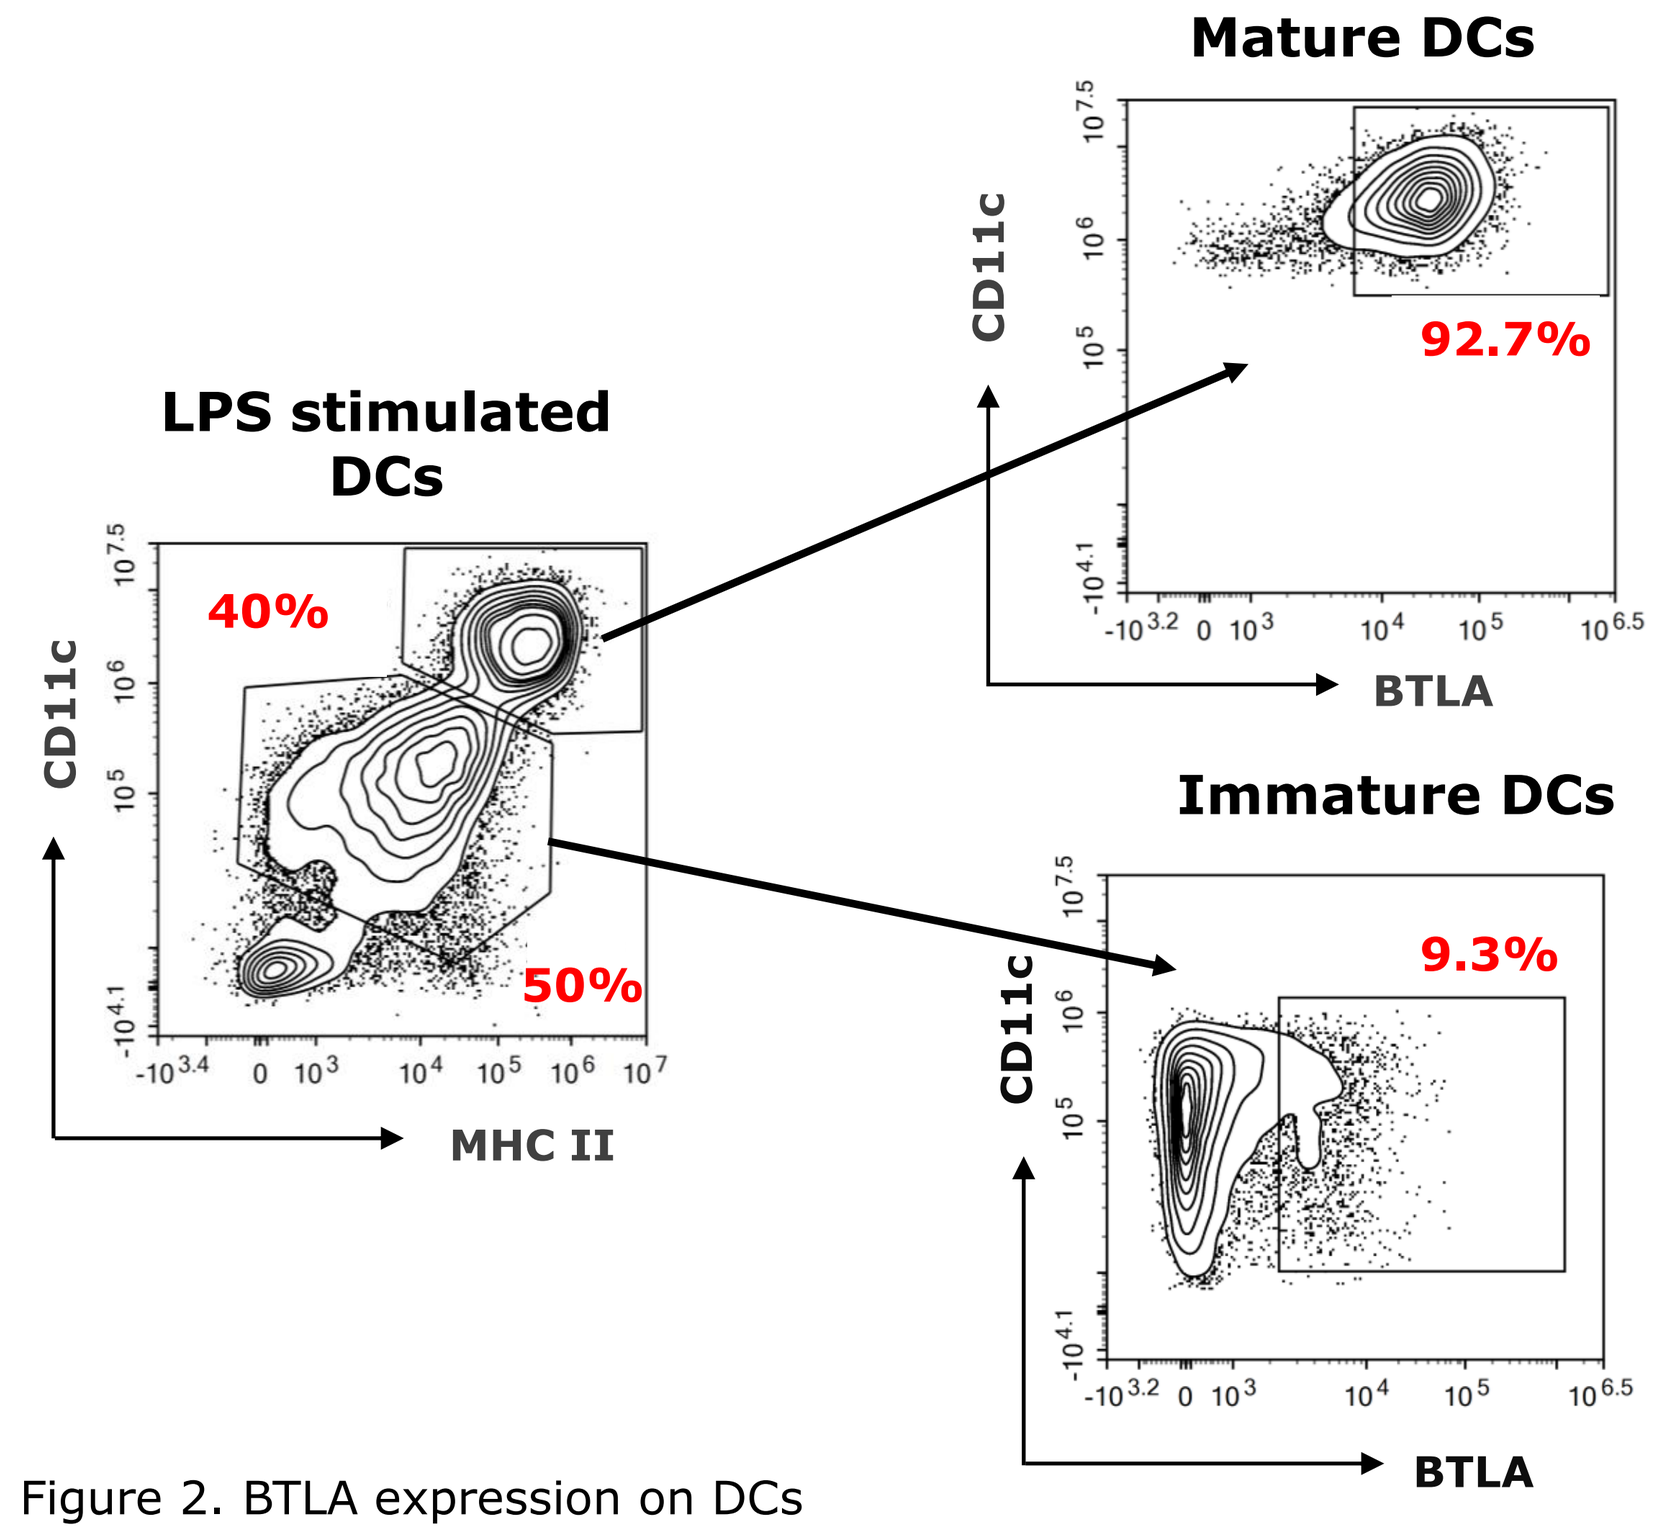
LPS stimulated DCs
40%
50%
CD11c
MHC II
Mature DCs
92.7%
CD11c
BTLA
Immature DCs
9.3%
CD11c
BTLA

Figure 2. BTLA expression on DCs

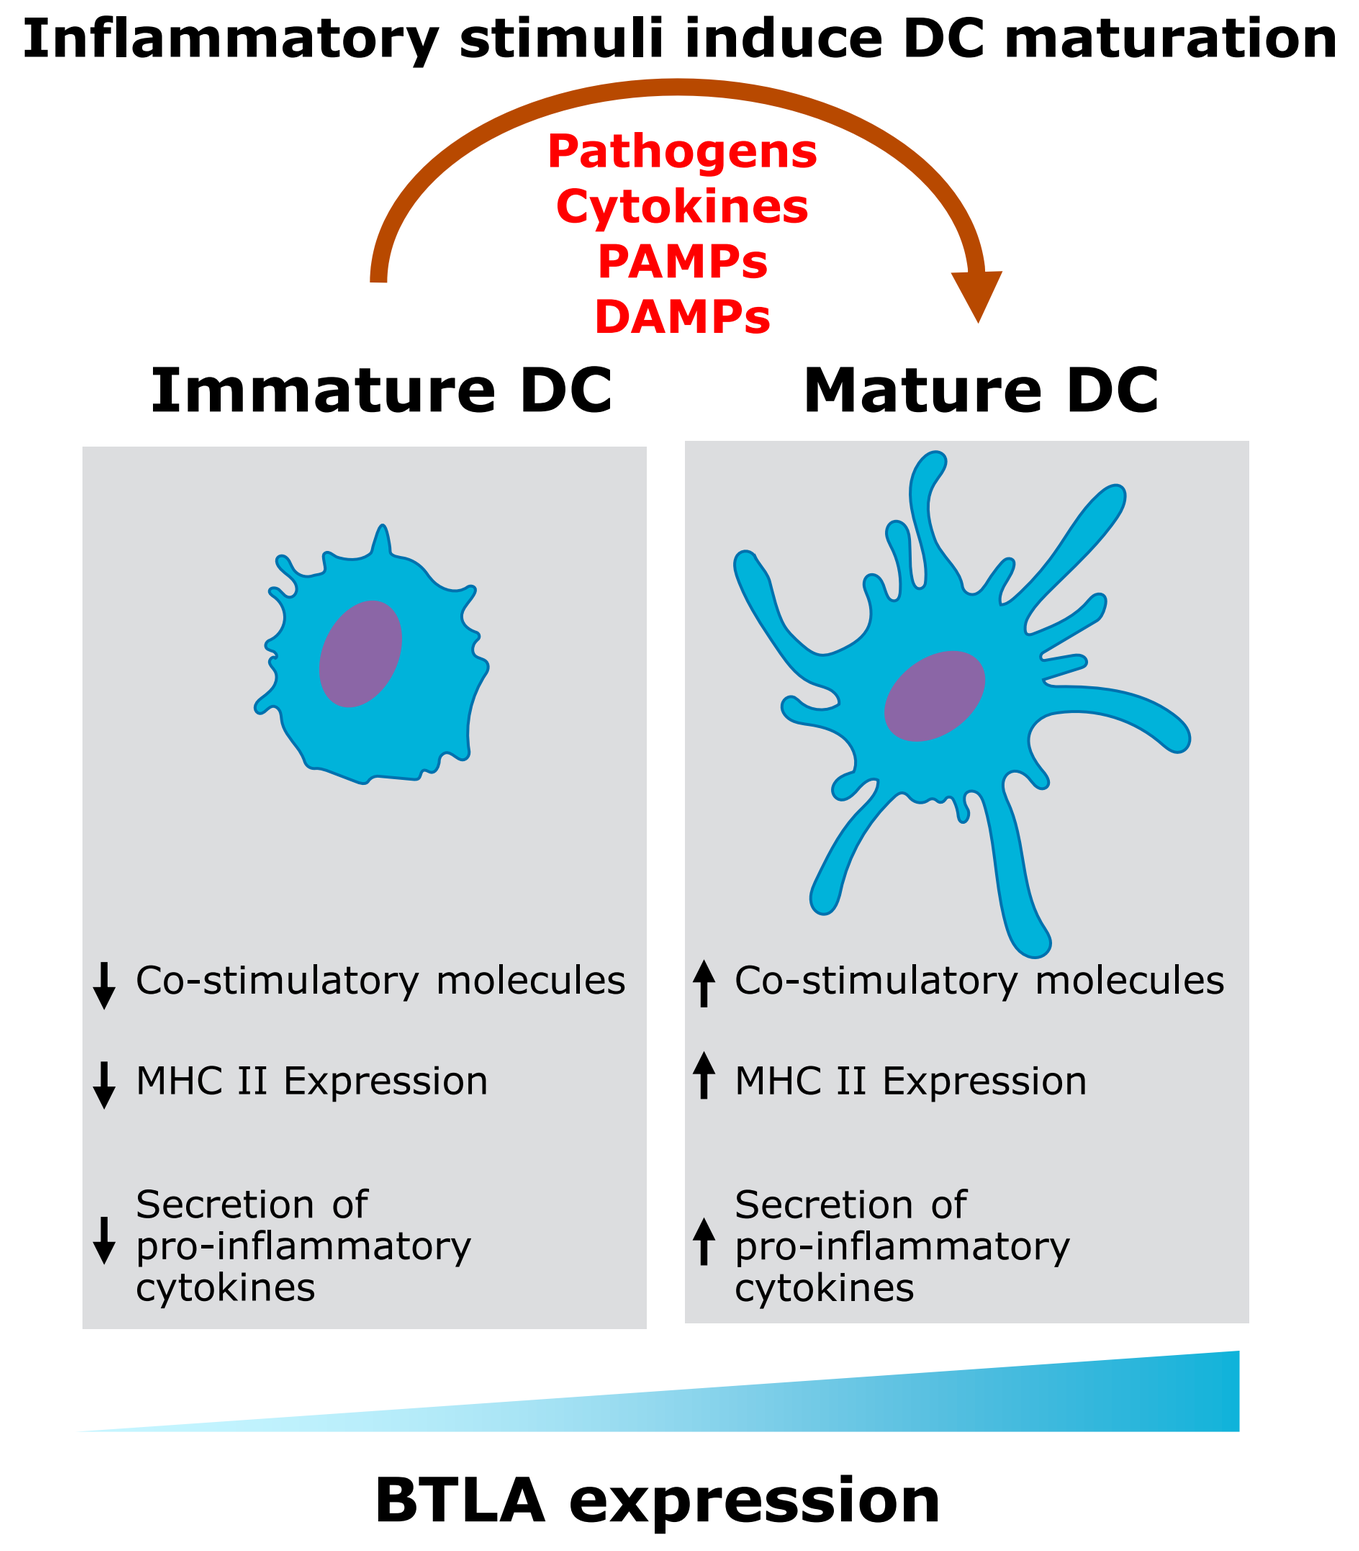
Inflammatory stimuli induce DC maturation
Pathogens
Cytokines
PAMPs
DAMPs
Immature DC
Mature DC
Co-stimulatory molecules
MHC II Expression
Secretion of pro-inflammatory cytokines
Co-stimulatory molecules
MHC II Expression
Secretion of pro-inflammatory cytokines
BTLA expression

Adapted from Patente et al, Front. Immunol., 2019

ANB032 Inhibits DC Maturation and Reduces Antigen Presentation and Co-Stimulatory Molecules

- After treatment with either ANB032 or the isotype control, maturation of both mature and immature LPS-stimulated DCs were inhibited and the absolute number of mature DCs was substantially less than control (**Figure 3A**)
- ANB032-treated DCs had lower expression of MHC II relative to the isotype control, indicating a reduction in antigen presentation with ANB032 (**Figure 3B**)
- ANB032 reduced expression of co-stimulatory molecules, CD80, CD86, and CD40, relative to the isotype control (**Figure 3C**)

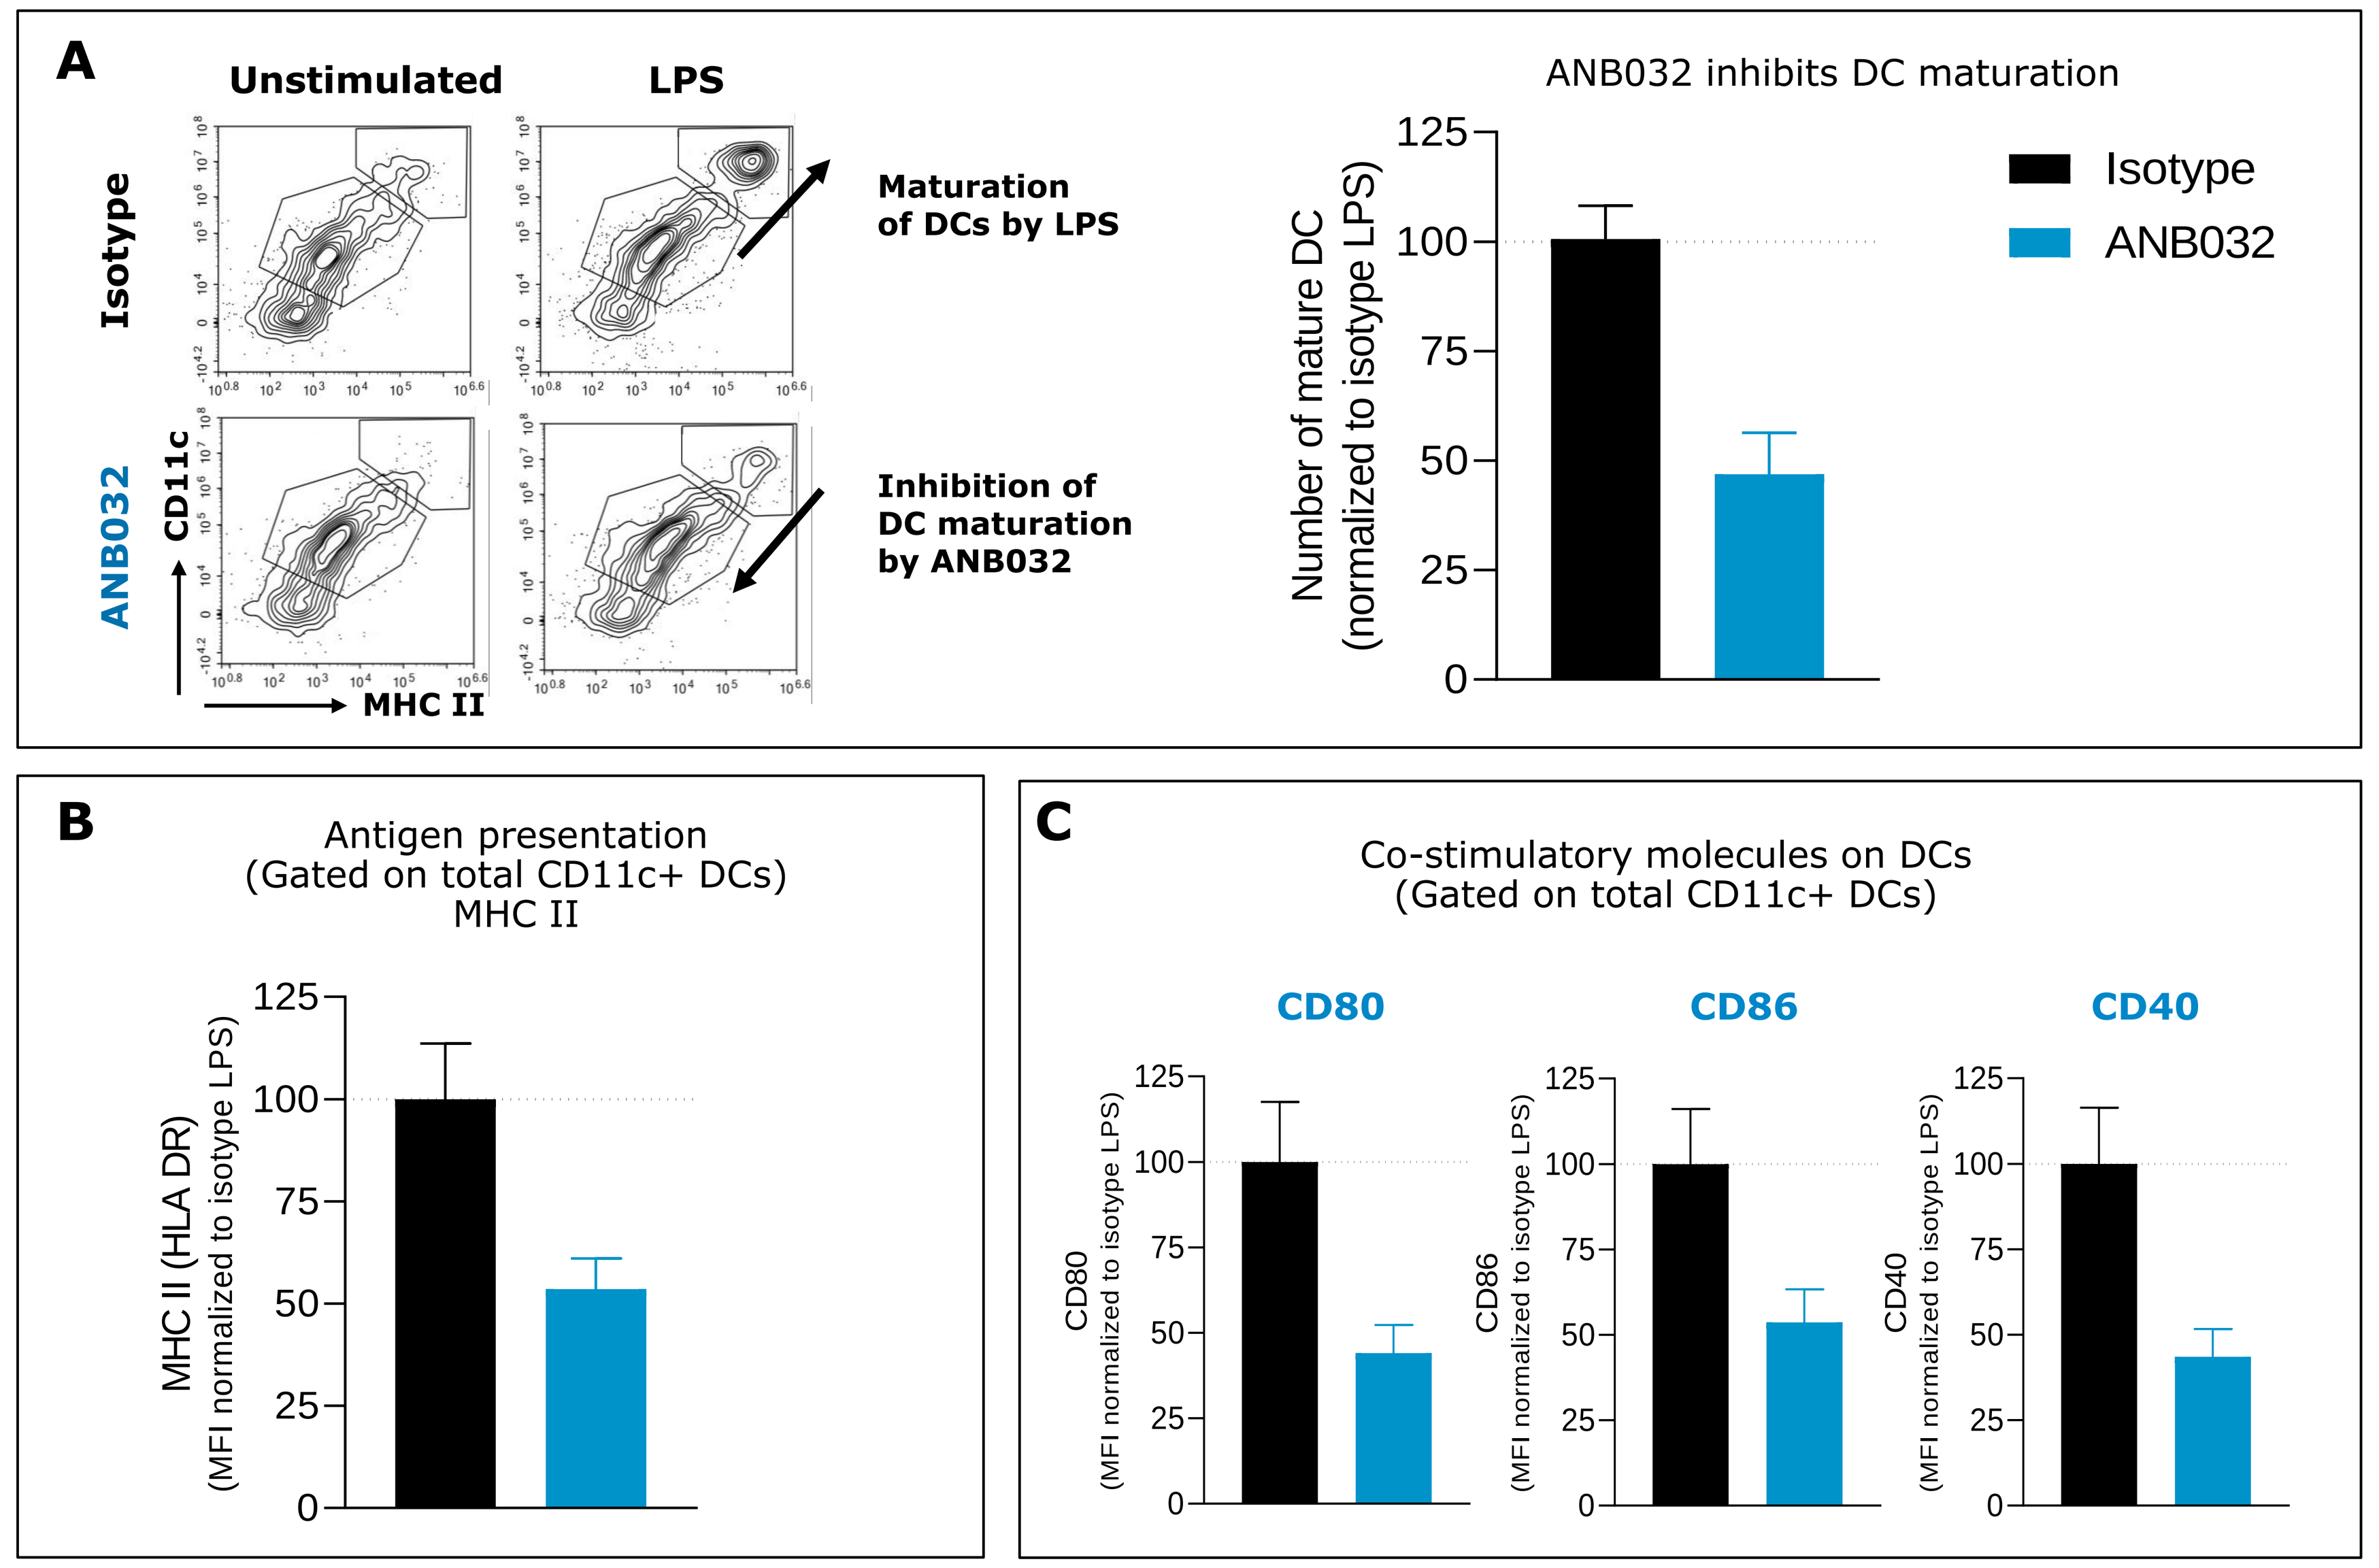
A
Unstimulated
LPS
Isotype
ANB032
CD11c
MHC II
Maturation of DCs by LPS
Inhibition of DC maturation by ANB032
ANB032 inhibits DC maturation
Number of mature DC (normalized to isotype LPS)
Isotype
ANB032
B
Antigen presentation
(Gated on total CD11c+ DCs)
MHC II
MHC II (HLA DR) (MFI normalized to isotype LPS)
C
Co-stimulatory molecules on DCs
(Gated on total CD11c+ DCs)
CD80
CD86
CD40
CD80 (MFI normalized to isotype LPS)
CD86 (MFI normalized to isotype LPS)
CD40 (MFI normalized to isotype LPS)

Figure 3. Effect of ANB032 on DCs

ANB032-treated DCs Induce Functional Tregs

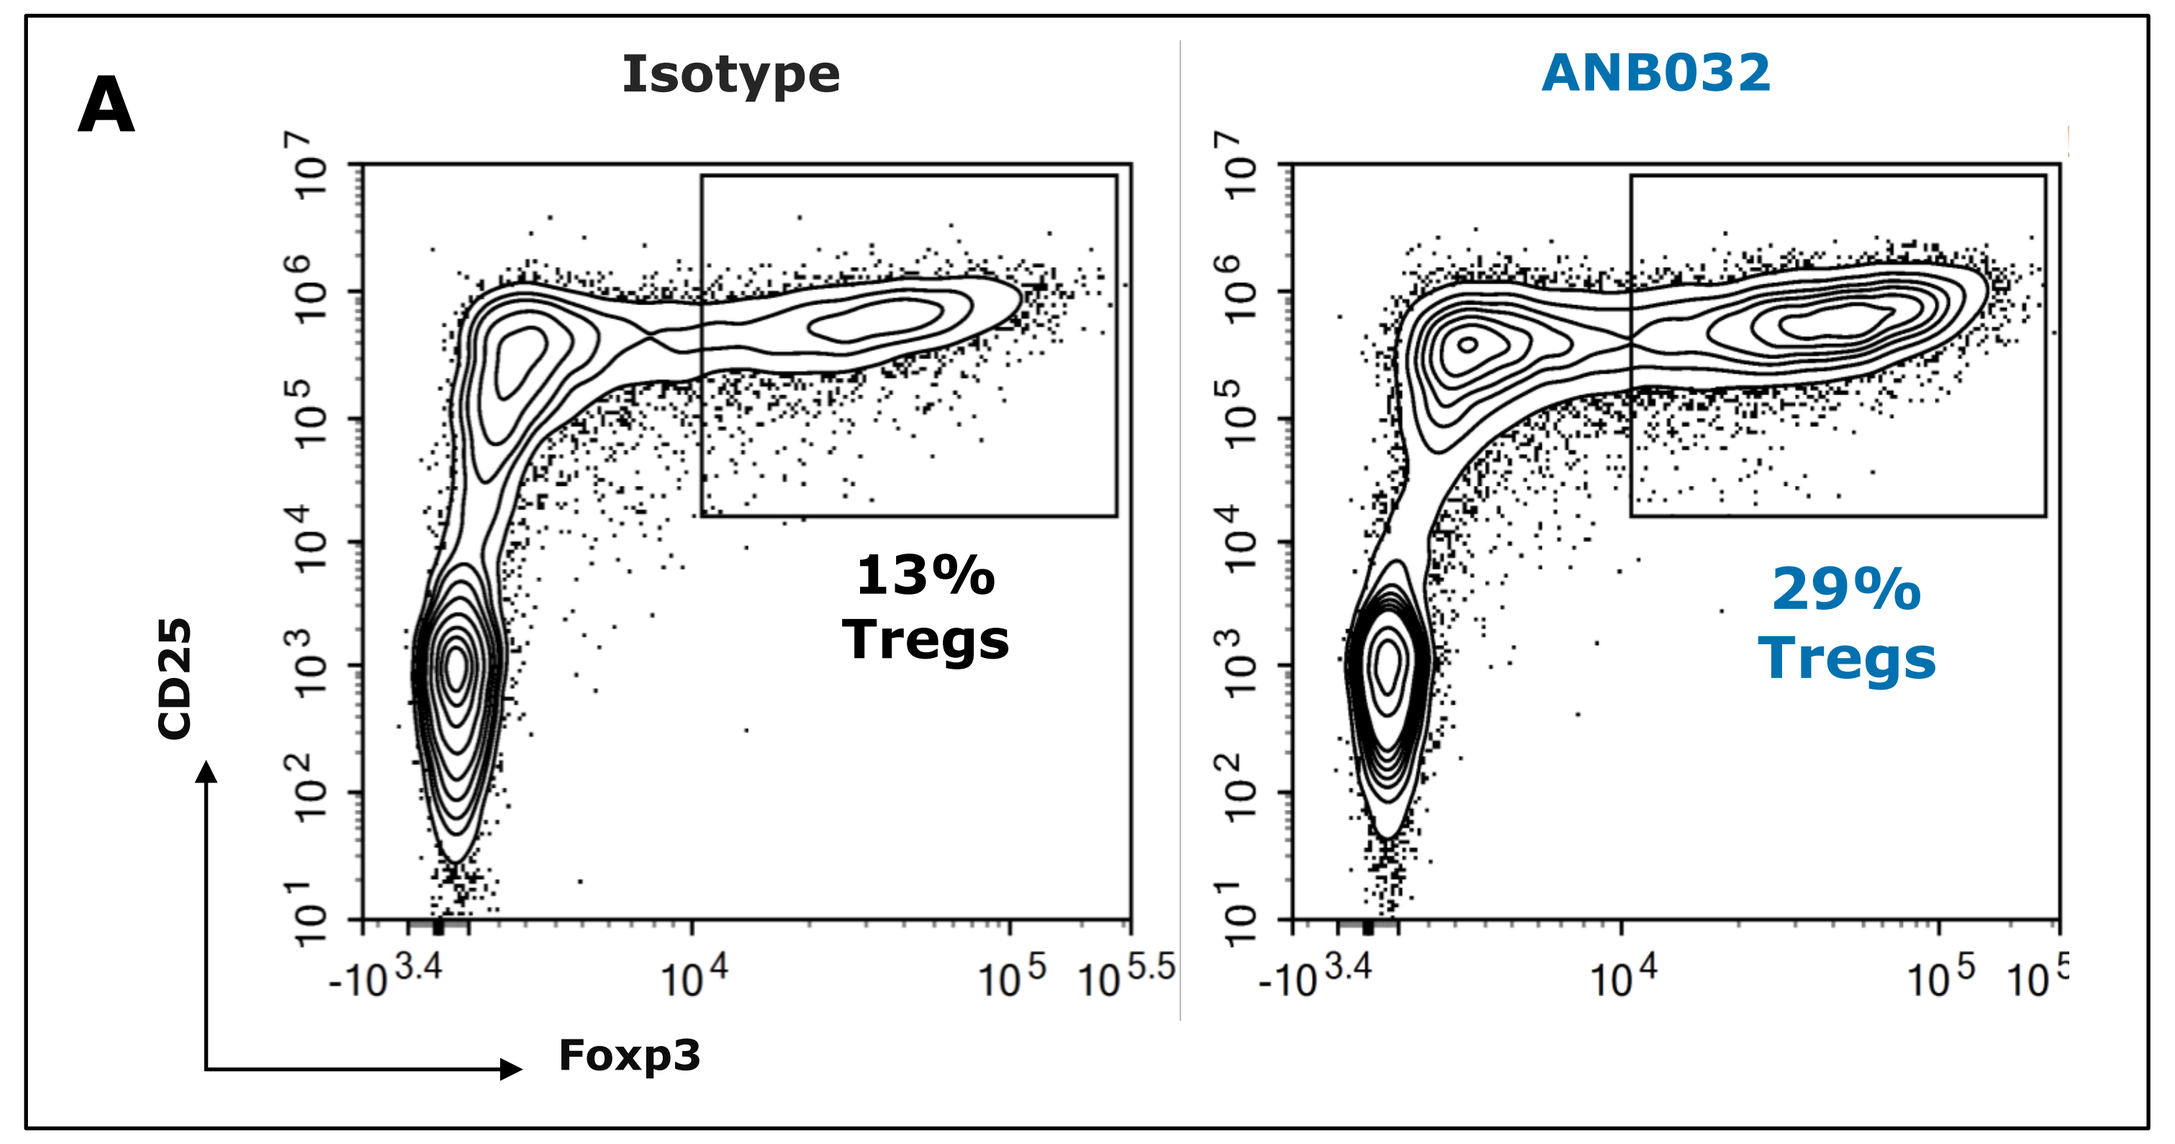
A
Isotype
ANB032
CD25
Foxp3
13% Tregs
29% Tregs

DCs were treated with either ANB032 or isotype and then co-cultured with allogenic naïve CD4 T cells to allow T cell differentiation. T cells were stained for CD4, CD25 and intracellular Foxp3 to identify inducible Tregs. (**Figure 4A**) The absolute number of Tregs was increased and inflammatory cytokines were reduced with ANB032 pre-treated DCs (**Figure 4B**).

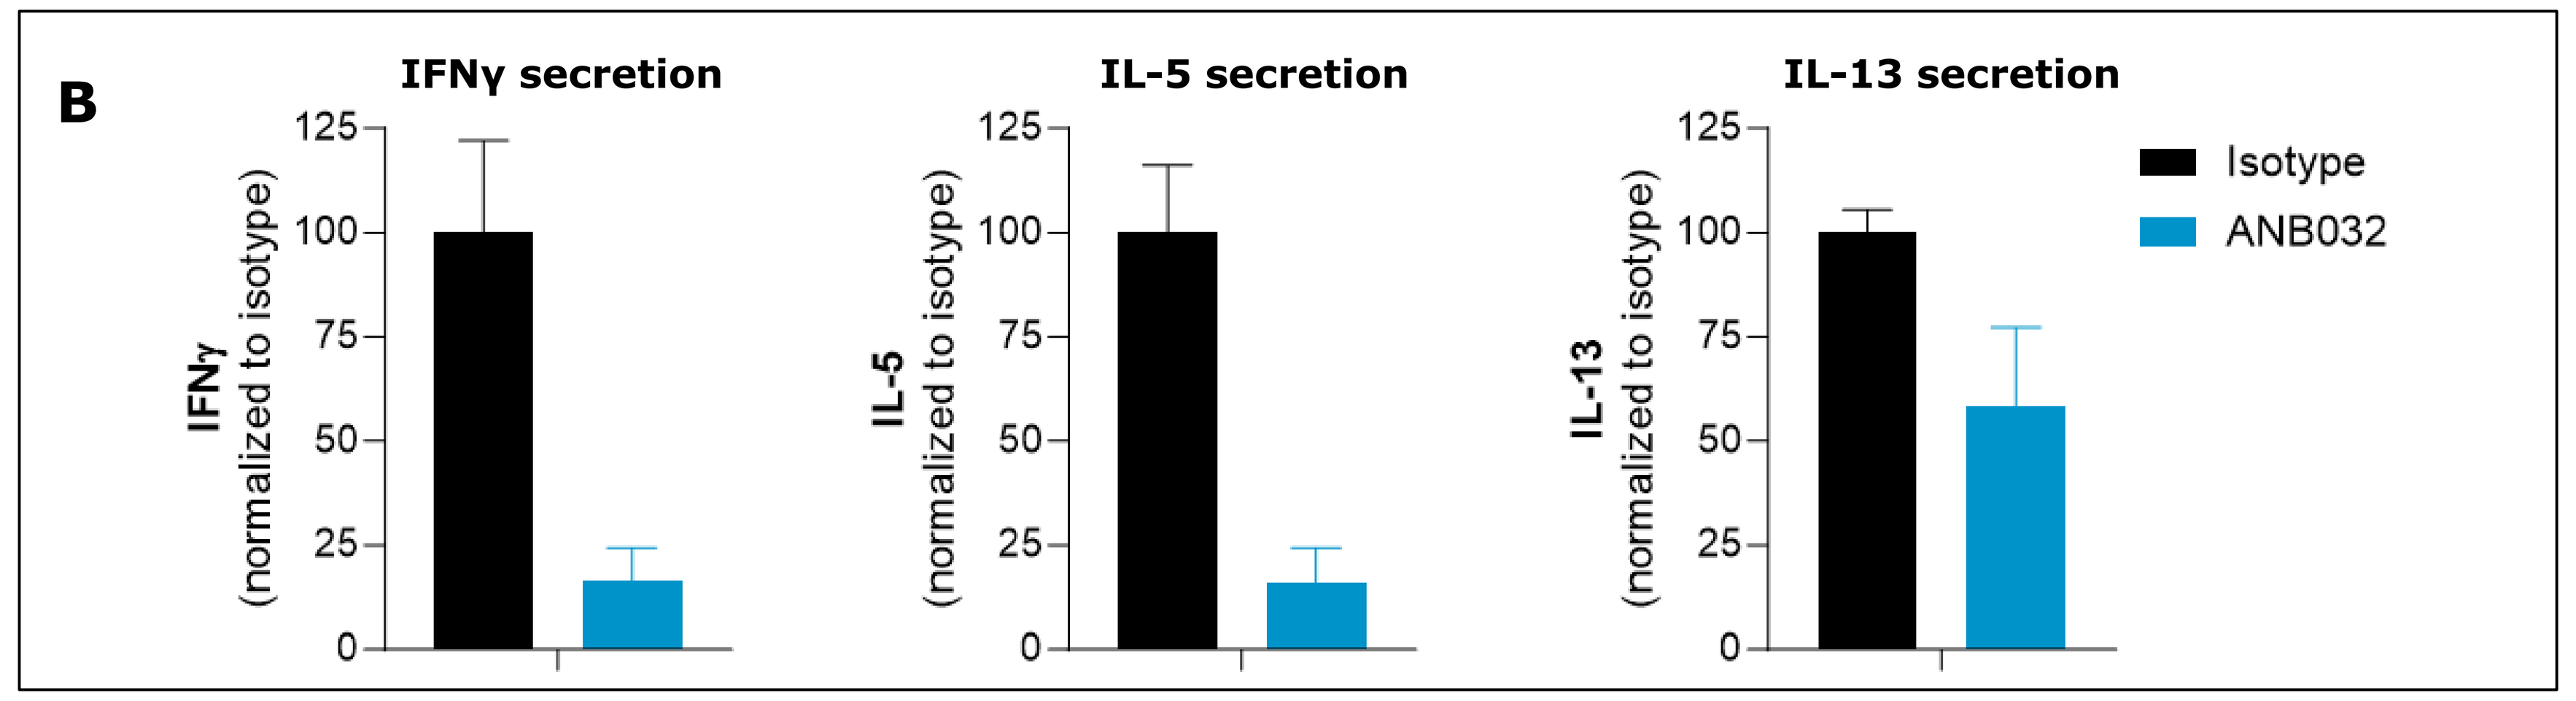
B
IFNγ secretion
IL-5 secretion
IL-13 secretion
IFNγ (normalized to isotype)
IL-5 (normalized to isotype)
IL-13 (normalized to isotype)
Isotype
ANB032

Figure 4. Effect of ANB032-treated DCs on functional Tregs and cytokine secretion

CONCLUSIONS

- BTLA is highly expressed on mature DCs
- Preclinical evaluation of ANB032 demonstrated:
 - Inhibition of DC maturation and reduction of antigen presentation and co-stimulatory molecule expression
 - Modulation of DC function to boost inducible Foxp3+ Tregs and inhibit effector T cells and inflammatory cytokine production
 - Inhibition of a broad range of immune cells, including DCs, while inducing Tregs, which supports a potential for restoring immune balance
- Based on these findings, ANB032 may provide therapeutic value in the treatment of autoimmune and inflammatory diseases, including AD
- A double-blind, placebo-controlled, global Phase 2 study of ANB032 in moderate-to-severe AD is actively enrolling subjects (NCT05935085)

ACKNOWLEDGEMENTS

- This research was funded by Anaptys
- Disclosures: All authors are employees and stockholders of Anaptys

REFERENCES

1. Luu, et al. Presented at ISID 2023
2. Guttman-Yassky, et al. *J Allergy Clin Immnol* 2007;119:1210-7.
3. Nakagomi et al. *J Invest Dermatol* 2013;133:702-11.
4. Bekiaris et al. *Immunity* 2013;39:1082-94.

Presented at the American Academy of Dermatology Annual Meeting, San Diego, CA, USA, March 8-12, 2024