Discovery of a PD-1 Checkpoint Agonist Antibody for Autoimmune/Inflammatory Disease

Marilyn R. Kehry, Gerald Manorek, Janean Fisher, Natasha Del Cid, Allison Rooks, Laurence Altobell III, Gregory Gold, Morena Shaw, Robert Morse, Margaret Marino, Jessie-Farah Fecteau, and Stephen Parmley. *AnaptysBio, Inc., San Diego, CA*

Introduction: Inhibitory Checkpoints Down-Modulate Immune Responses

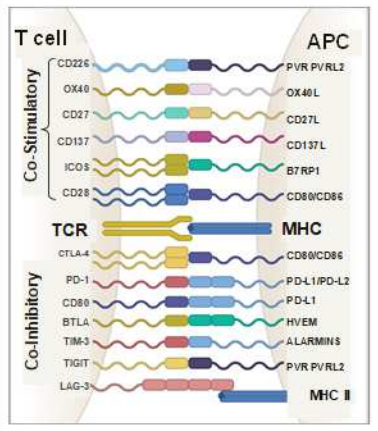
T cell
APC
Co-Stimulatory
CD226
OX40
CD27
CD137
ICOS
CD28
PVR PVRL2
OX40L
CD27L
CD137L
B7RP1
CD80/CD86
TCR
MHC
Co-Inhibitory
CTLA-4
PD-1
CD80
BTLA
TIM-3
TIGIT
LAG-3
CD80/CD86
PD-L1/PD-L2
PD-L1
HVEM
ALARMINS
PVR PVRL2
MHC II

- Immune checkpoint receptor-ligand interactions are essential for down-regulating immune responses and maintaining self-tolerance
- Functional antagonist antibodies to PD-1 and CTLA-4, major checkpoints on activated T cells, enhance existing immune responses and are approved therapeutics in multiple oncology indications
- Genetic mutations in the PD-1 pathway have been shown to increase susceptibility to various autoimmune and inflammatory diseases*
- We hypothesize that many human autoimmune diseases occur due to dysregulated PD-1 signaling, leading to uncontrolled T cell responses
- Agonist antibodies to PD-1 that mimic the function of natural ligands and augment PD-1 signaling have the potential to suppress human autoimmune/inflammatory diseases and reinstate tolerance

*Okazaki T and Honjo T (2007) *Int. Immunol.* **19**:813.

Anti-PD-1 Agonist Antibody Discovery

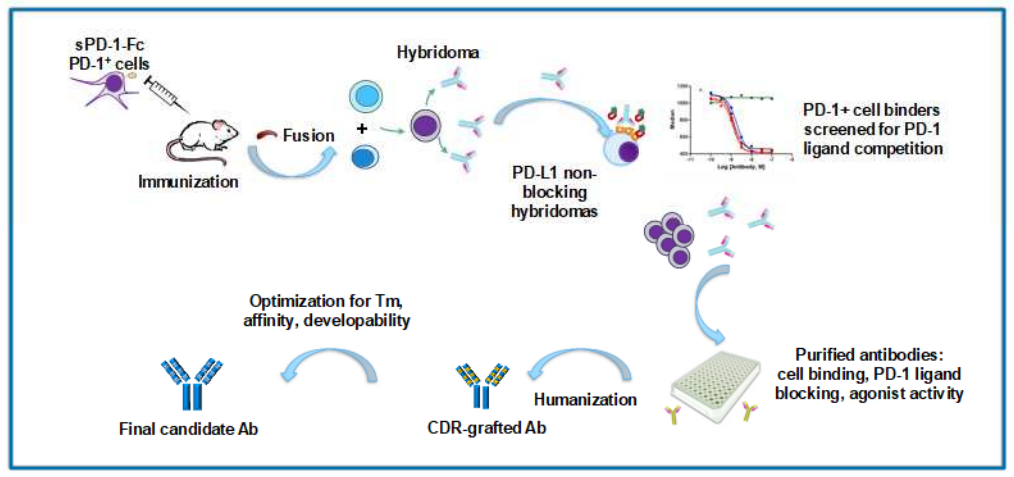
sPD-1-Fc
PD-1+ cells
Immunization
Fusion
Hybridoma
PD-L1 non-blocking hybridomas
PD-1+ cell binders screened for PD-1 ligand competition
Purified antibodies: cell binding, PD-1 ligand blocking, agonist activity
Humanization
CDR-grafted Ab
Optimization for Tm, affinity, developability
Final candidate Ab

ANB030 is a humanized IgG1,κ hybridoma-derived anti-PD-1 agonist antibody that was optimized for affinity and functional activity. It is PD-L1 non-blocking and lacks checkpoint antagonist activity. It has no observed ADCC activity on cells expressing native activated T cell levels of PD-1.

ANB030 Thermal Stabilization and Affinity Optimization

ANB030 Variant Antibody	K_D Human PD-1 (nM)	K_D Cynomolgus PD-1 (nM)	Fab Tm (°C)
Mouse chimeric	4.3	n.d.	n.d.
CDR-grafted	0.186	1.02 ± 0.08 N=2	61.2 ± 0.3 N=3
CDR-grafted Optimized (ANB030)	0.060	0.64 ± 0.10 N=2	66.1

K_D measurements for screening by Surface Plasmon Resonance (SPR) were performed on a Biacore T200, and kinetic constants were fit globally using a 1:1 binding model. Biotinylated human or cynomolgus monkey PD-1 extracellular domain monomer was captured at a 1 nM concentration on a Biacore Sensor chip SA with a carboxymethylated dextran surface pre-immobilized with streptavidin. The captured antigen level was targeted to yield a low response to prevent avidity effects on the dissociation rate. Tm measurements were determined by fluorescence-based thermal shift and differential scanning calorimetry.

ANB030 was additionally de-risked for pre-clinical development and manufacturing by assessing 28-day stability in human and cynomolgus monkey serum at 37°C, stability to freeze-thaw and low pH, pre-formulation, and stability at 100 mg/ml.

ANB030 Binding to Human and Cynomolgus PD-1 is High Affinity

KinExA
Human PD-1

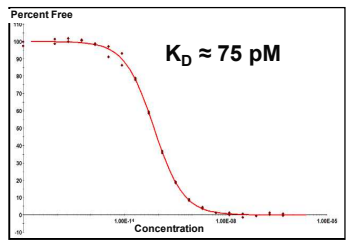
Percent Free
K_D ≈ 75 pM
Concentration

KinExA
Cynomolgus Monkey

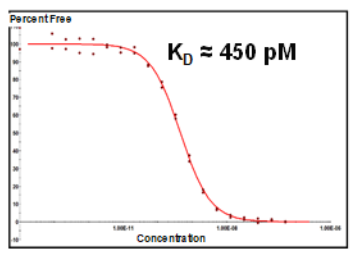
Percent Free
K_D ≈ 450 pM
Concentration

Solution-based Kinetic Exclusion Assay affinity measurements for ANB030 binding human and cynomolgus monkey PD-1 were determined on a KinExA 3000. ANB030 was incubated with a broad concentration range (500 nM – 86 fM) of soluble human or cynomolgus monkey PD-1 monomer to establish equilibrium binding, and free antibody was captured on azlactone beads coated with either human or cynomolgus monkey PD-1 monomer. Detection of ANB030 bound to the beads was with Alexa Fluor 647 anti-human IgG. K_D values were determined using a 1:1 reversible binding interaction model.

ANB030 Binding to PD-1 Does Not Compete with PD-L1 Binding

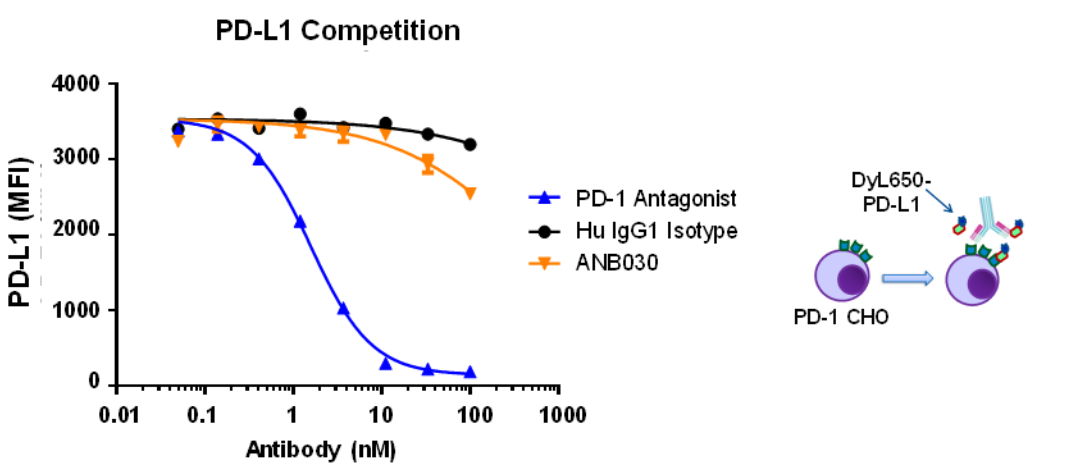
PD-L1 Competition
PD-L1 (MFI)
4000
3000
2000
1000
0
0.01
0.1
1
10
100
1000
Antibody (nM)
PD-1 Antagonist
Hu IgG1 Isotype
ANB030
DyL650-PD-L1
PD-1 CHO

Human PD-L1-mIgG1 Fc labeled with DyLight 650 was mixed with various concentrations of anti-PD-1 antibody or isotype control antibody. The mixture was added to human PD-1 transfected CHO-K1 cells, incubated to allow PD-L1 and anti-PD-1 binding, washed, and the fluorescence of bound PD-L1-DyLight 650 was quantified by flow cytometry on a BD FACSArray.

ANB030 Inhibits CD4⁺ T Cell Cytokine Production

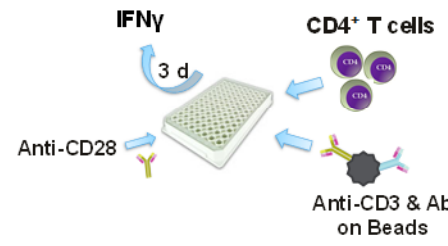
IFNγ
3 d
CD4⁺ T cells
Anti-CD28
Anti-CD3 & Ab on Beads

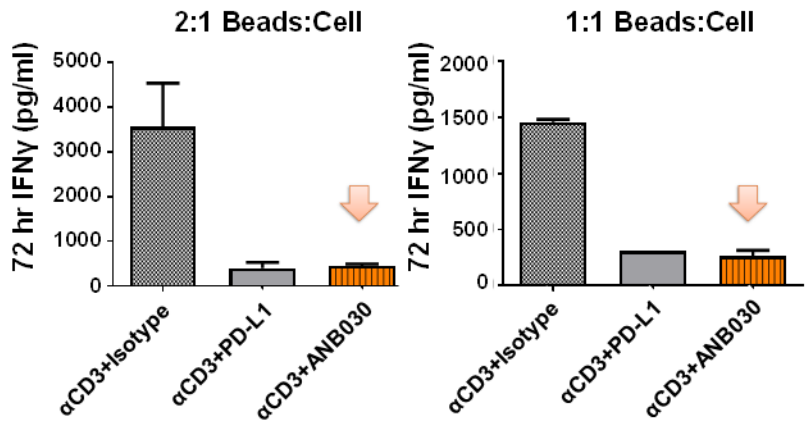
2:1 Beads:Cell
1:1 Beads:Cell
72 hr IFNγ (pg/ml)
5000
4000
3000
2000
1000
0
2000
1500
1000
500
0
αCD3+Isotype
αCD3+PD-L1
αCD3+ANB030
Protein Coupled Beads

Antibody	% IFNγ Inhibition (Mean ± SEM)	Donors (N)
ANB030	77 ± 7	6

Anti-CD3 and either ANB030, PD-L1-Fc, or a human IgG1 isotype control antibody were coupled to M280 magnetic beads. Consistent levels of anti-CD3 coupling across bead lots were confirmed by flow cytometry. Human CD4+ T cells freshly purified from normal donor blood were incubated with different bead:cell ratios in the presence of 250 ng/ml soluble anti-CD28 for 72 hours. Secretion of IFNγ in the culture supernatants was quantified by ELISA. Bead:Cell ratio was used to determine T cell activation window for responsiveness to PD-L1.

ANB030 Shows Well-Behaved Pharmacokinetic Properties in Cynomolgus Monkeys with Good Bioavailability after Subcutaneous Dosing

ANB030 Single Dose Pharmacokinetics

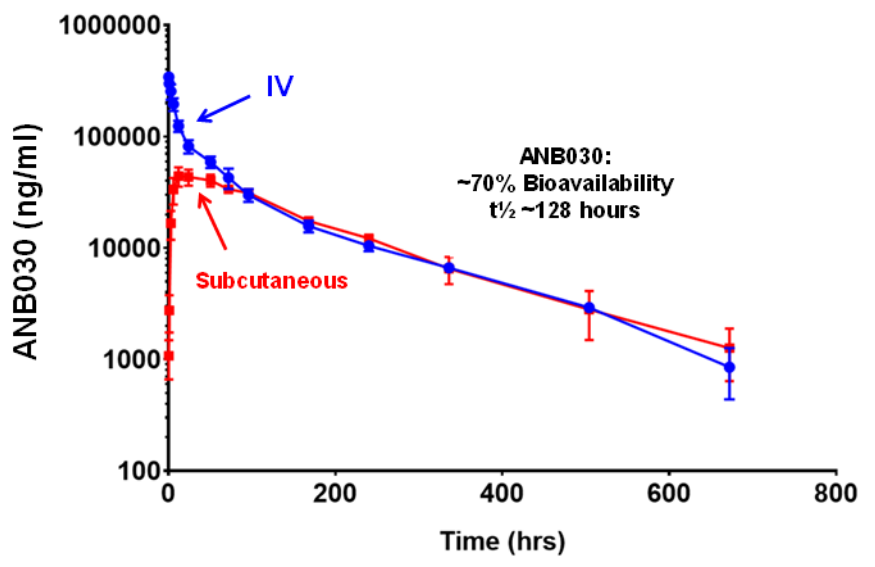
ANB030 (ng/ml)
1000000
100000
10000
1000
100
0
200
400
600
800
Time (hrs)
IV
Subcutaneous
ANB030:
~70% Bioavailability
t½ ~128 hours

A single 10 mg/kg dose of ANB030 was administered either intravenously or subcutaneously to biologics naïve male cynomolgus monkeys. ANB030 in serum samples taken at various times after dosing was quantified by ELISA. Each point is the mean serum concentration of ANB030 of 3 animals/group.

ANB030 in Solution Potently Inhibits Tetanus Toxoid Recall Response in Whole Blood: Functional Activity is Dependent on FcγR Engagement

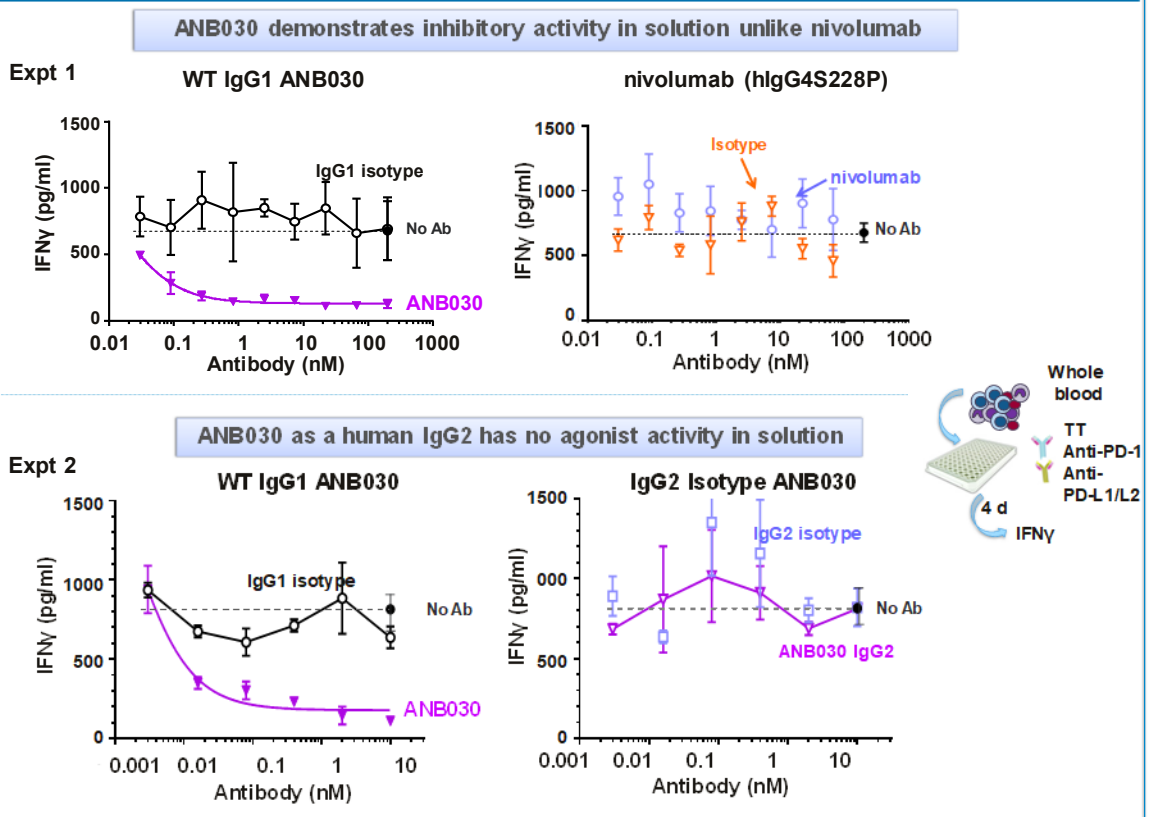
ANB030 demonstrates inhibitory activity in solution unlike nivolumab
Expt 1
WT IgG1 ANB030
nivolumab (hIgG4S228P)
IFNγ (pg/ml)
IgG1 isotype
No Ab
ANB030
Isotype
nivolumab
Antibody (nM)
ANB030 as a human IgG2 has no agonist activity in solution
Expt 2
WT IgG1 ANB030
IgG2 Isotype ANB030
IgG2 isotype
ANB030 IgG2
Whole blood
TT
Anti-PD-1
Anti-PD-L1/L2
4 d
IFNγ

Human whole blood from tetanus toxoid responsive donors was incubated at 37°C in the presence of tetanus toxoid (TT, 5 µg/ml), anti-PD-L1 and anti-PD-L2 (2 µg/ml each), and various concentrations of ANB030, human IgG1 isotype control, or nivolumab. After 4 days plates were centrifuged, and IFNγ in plasma was quantified by ELISA. Each point is the mean ± SEM of 4 replicate wells.
ANB030 as a human IgG2 had identical activated T cell binding as ANB030 IgG1. The lack of agonist activity of ANB030 IgG2 (shown above), IgG4, or IgG1(L234A,L235A) isotypes (results not shown) demonstrates a requirement for FcγR engagement/ANB030 clustering for functional agonist activity.

ANB030 Induces SHP2 but not SHP1 Recruitment to PD-1 after Activation of Jurkat PD-1 Cells

PD-1 Signaling in T Cells

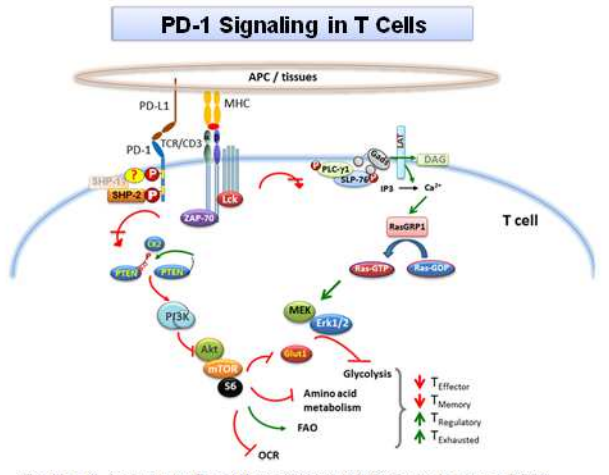
APC / tissues
PD-L1
MHC
PD-1
TCR/CD3
SHP-2
Lck
ZAP-70
PLC-γ1
SLP-76
LAT
DAG
IP3
RasGRP1
T cell
PTEN
PI3K
Akt
mTOR
S6
MEK
Erk1/2
Glut1
Glycolysis
Amino acid metabolism
FAO
OCR

Bardhan K, Anagnostou T and Boussiotis VA (2016) *Front. Immunol.* 7:550.

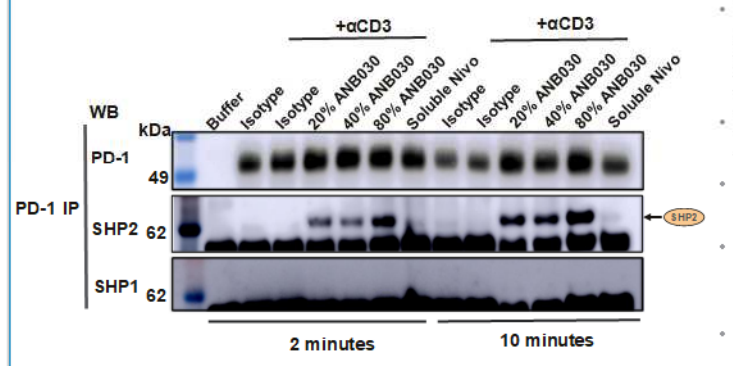
+αCD3
+αCD3
WB
kDa
Buffer
Isotype
Isotype
20% ANB030
40% ANB030
80% ANB030
Soluble Nivo
Isotype
Isotype
20% ANB030
40% ANB030
80% ANB030
Soluble Nivo
PD-1 IP
PD-1
49
SHP2
62
SHP1
62
SHP2
2 minutes
10 minutes

- In combination with T-cell activation, ANB030 induced recruitment of SHP2 but not SHP1 to PD-1, consistent with published literature on PD-1 signaling
- Increased SHP2 recruitment was found with higher density ANB030 on the beads
- No SHP recruitment was found with soluble nivolumab
- In combination with T-cell activation and CD28 co-stimulation, ANB030 also reduced ZAP70 and LAT phosphorylation (not shown)
- ANB030 had no effect on signaling pathways in the absence of T-cell activation (not shown)

Jurkat PD-1 cells were activated with anti-CD3 and ANB030 on beads to mimic FcγR-dependent binding of ANB030 to antigen presenting cells. Nivolumab is a human IgG4(S228P) antibody and was added in solution to reflect its lack of FcγR binding. After the indicated stimulation times cells were lysed, PD-1 was immunoprecipitated, and immunoprecipitates subjected to SDS-PAGE. Immunoblots were probed with anti-PD-1, anti-SHP2, or anti-SHP1 antibodies.

Conclusions

- **Functional agonist anti-PD-1 antibodies that down-regulate antigen-specific immune responses and lack antagonist activity can be discovered and optimized**
- **ANB030 is a humanized IgG1/κ anti-PD-1 agonist antibody that is non-blocking for PD-L1 binding and requires Fcγ receptor engagement for its functional inhibitory activity in solution**
- **ANB030 has been de-risked for pre-clinical development: ANB030 is high affinity, has been optimized for stability, and shows good bioavailability and pharmacokinetics using subcutaneous dosing in cynomolgus monkeys**
- **ANB030 demonstrated efficacy in a xenogeneic NSG-Human-PBMC graft vs. host disease model (not shown)**
- **Signaling mechanism studies show similar PD-1-dependent effects for ANB030 and PD-L1-Fc: T cell activation-dependent recruitment of SHP2 but not SHP1 to PD-1 and inhibition of key T cell activation-dependent signaling pathways (phospho-ZAP70 and phospho-LAT)**
- **Anti-PD-1 antibodies that mimic activity of natural checkpoint ligands and down-modulate T-cell responses have the potential to restore and maintain immune balance in autoimmune and inflammatory diseases**