Discovery and Characterization of ANB032, a Novel BTLA/HVEM Checkpoint Modulator for Autoimmune/Inflammatory Disease

Martin E. Dahl, Jean da Silva, Gerald Manorek, Natasha Del Cid, April Fraley, Gregory Gold, Robert Morse, Margaret Marino, JingUei Verkade, Yu-yu Ren, Janean Young, Paul Fisher, Stephen Parmley and Marilyn R. Kehry. *AnaptysBio, Inc., San Diego, CA*

Abstract

Human genetics and animal studies demonstrate a role for B and T lymphocyte attenuator (BTLA) in autoimmune disease. BTLA is an immune checkpoint molecule that contributes to the regulation of T cell, B cell and dendritic cell function through its interaction with its ligand herpesvirus entry mediator (HVEM) on cells in *trans*, and on the same cell in *cis*. *Trans* BTLA/HVEM interaction results in inflammatory costimulatory signaling through HVEM, while *cis* BTLA/HVEM interaction prevents costimulatory signaling of HVEM by numerous HVEM ligands, resulting in broad suppression of HVEM ligand-mediated immune cell activity.

We have discovered a novel anti-BTLA monoclonal antibody, ANB032, that preserves the anti-inflammatory BTLA/HVEM *cis* complex formation and blocks the pro-inflammatory co-stimulatory signal mediated through HVEM by *trans* BTLA binding. ANB032 also demonstrated the potential to directly induce co-inhibitory agonistic signaling through BTLA, which was potentiated by Fc receptor engagement. Hydrogen-deuterium exchange mapping of the ANB032 binding epitope on BTLA demonstrated that ANB032 bound a region distal from the HVEM binding site, providing evidence to support the lack of disruption of the BTLA/HVEM complex by ANB032.

***In vivo*, ANB032 was highly efficacious in a dose dependent manner when tested in a human PBMC-NOD-*scid IL2rγ*[null] (NSG) graft versus host disease model, while an anti-BTLA antibody that antagonized the BTLA/HVEM interaction had no therapeutic effect. Thus, anti-BTLA antibodies that regulate BTLA/HVEM interactions to down-modulate T cell responses have the potential to restore and maintain immune balance in autoimmune and inflammatory diseases.**

Introduction: Bi-directional *trans* BTLA-HVEM interactions determine functional biology and can be modulated by ANB032

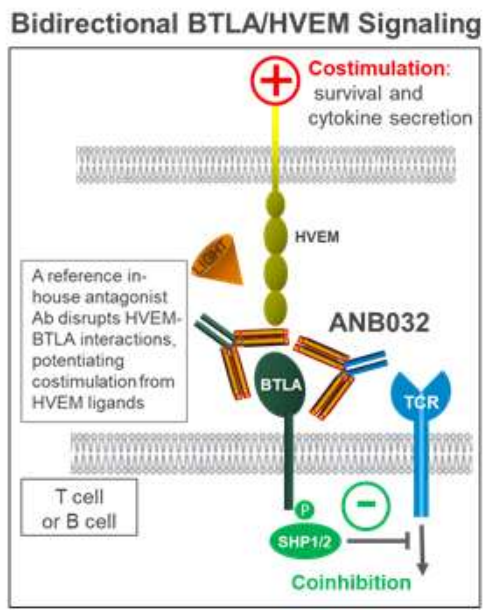
Bidirectional BTLA/HVEM Signaling
Costimulation: survival and cytokine secretion
HVEM
LIGHT
A reference in-house antagonist Ab disrupts HVEM-BTLA interactions, potentiating costimulation from HVEM ligands
ANB032
BTLA
TCR
T cell or B cell
P
SHP1/2
Coinhibition

BTLA was first identified as a negative immune regulator on T and B cells, most closely related to PD-1 and CTLA-4. In contrast to other inhibitory receptors whose expression is induced after cell activation, BTLA is constitutively expressed by many immune cells (Šedý et al., 2005; Watanabe et al., 2003). BTLA expression levels vary substantially among different lymphoid and myeloid subtypes with highest expression on B cells but also significant expression on T cells as well as on mature dendritic cells, macrophages and natural killer cells (Han et al., 2004; Hurchla et al., 2005).

The ligand of BTLA is Herpes-Virus entry mediator (HVEM). In the past decade, HVEM has emerged as a major and complex co-stimulatory signaling molecule. In addition to binding to BTLA, HVEM also serves as a receptor for four other ligands: LIGHT, lymphotoxin-a (Mauri et al., 1998), glycoprotein D (HSV-1 gD) (Spear et al., 2004; Murphy et al., 2008), and CD160 (Cai, et al., 2008; del Rio et al., 2010).

Engagement of BTLA by HVEM in *trans* across two cells induces Tyr phosphorylation of the ITIM motifs in BTLA, allowing the recruitment of the Src-homology domain 2-containing protein tyrosine phosphatases (SHP1 and SHP2) (Gavrieli et al., 2003; Vendel et al., 2009). Recruitment of SHP1 and SHP2 results in the attenuation of T cell proliferation, growth and cytokine production, consistent with an inhibitory function for BTLA. Conversely, HVEM as a receptor, instead of a ligand, mediates proinflammatory signaling in T cells (Cai et al., 2009). Engagement of HVEM via ligands such as LIGHT provide a co-stimulatory signal on T cells and B cells (Murphy et al., 2010; Duhen et al., 2004; Morel et al., 2001). HVEM and BTLA interactions can also occur in *cis* on the same cell, resulting in BTLA-mediated silencing of co-stimulatory signals through HVEM. Stabilization of this *cis* complex is thought to be one of the mechanisms by which BTLA regulates HVEM costimulation.

The complexity of the BTLA/HVEM network makes therapeutic inhibition strategies more challenging than for other inhibitory receptors or ligands. Thus, BTLA is emerging as a unique immune checkpoint receptor and interesting therapeutic target in cancer and autoimmunity. We report an anti-human BTLA antibody designated ANB032 that modulates the BTLA/HVEM bidirectional signaling pathways via three potential mechanisms: 1) ANB032 does not compete with BTLA/HVEM interactions and thus does not prevent HVEM-mediated *trans* inhibitory signaling through BTLA. However binding of ANB032 does inhibit BTLA-mediated co-stimulatory, pro-inflammatory signaling through HVEM in *trans*. 2) ANB032 permits BTLA/HVEM *cis* complex formation, inhibiting co-stimulation mediated through HVEM ligands. 3) ANB032 has the capacity to directly induce inhibitory agonistic signaling through BTLA, which is potentiated by Fc receptor engagement. Via these mechanisms, ANB032 has potential to restore and maintain immune balance in autoimmune and inflammatory diseases.

Figure 1: ANB032 preserves HVEM binding to BTLA

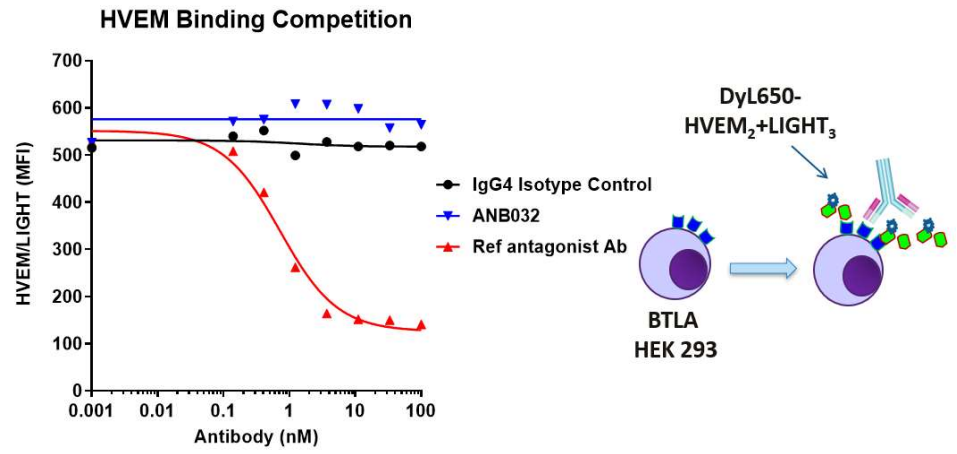
HVEM Binding Competition
HVEM/LIGHT (MFI)
700
600
500
400
300
200
100
0
0.001
0.01
0.1
1
10
100
Antibody (nM)
IgG4 Isotype Control
ANB032
Ref antagonist Ab
DyL650-HVEM$_2$+LIGHT$_3$
BTLA HEK 293

ANB032 does not compete with HVEM/LIGHT binding to BTLA expressed on 293c18 cells. A concentration titration of ANB032, isotype, or reference antagonist were preincubated on BTLA-expressing cells at concentrations indicated. DyL650-HVEM/LIGHT complexes (final concentration of HVEM/LIGHT, 30 nM each) were added for 30 min on ice then analyzed by flow cytometry. The ref antagonist antibody prevented HVEM binding while ANB032 did not, similar to the isotype control.

Figure 2: HD exchange mapping of ANB032 epitope on BTLA

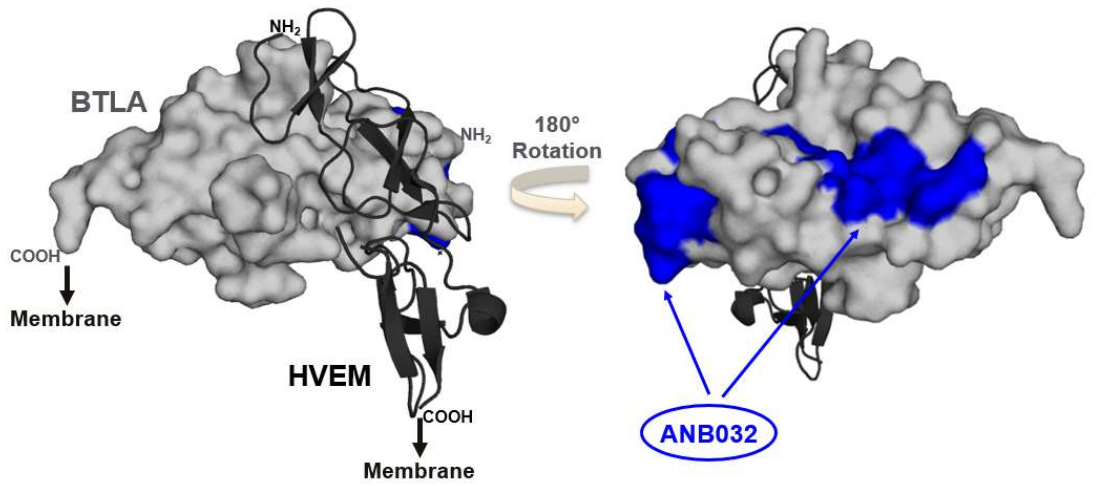
BTLA
NH_2
NH_2
COOH
Membrane
HVEM
COOH
Membrane
180° Rotation
ANB032

Hydrogen-deuterium (HD) exchange mass spectrometry was utilized for epitope mapping the binding regions of ANB032 on BTLA. Monomeric huBLTA-ECD-his protein was utilized which consisted of BTLA amino acids 31-157, followed by a C-terminal histidine tag. An HD exchange experiment was performed with ANB032 or reference antagonist antibody. Using the BTLA/ANB032 complex, two BTLA peptides (residues 52-65 and 100-106) were protected from HD exchange by ANB032. The HD exchange results were modeled on the three dimensional structure of BTLA in complex with HVEM. The BTLA peptides protected from HD exchange by ANB032 were found to be located on the opposite face of BTLA relative to the HVEM binding site. In contrast, peptides protected by the reference antagonist antibody were adjacent to or buried in the BTLA/HVEM binding interface (data not shown). These results provide a high resolution characterization of the ANB032 binding epitope on BTLA and are consistent with the functional data showing that ANB032 is not a direct antagonist of the BTLA-HVEM interaction.

Figure 3: ANB032 inhibits HVEM signaling induced by BTLA in *trans*

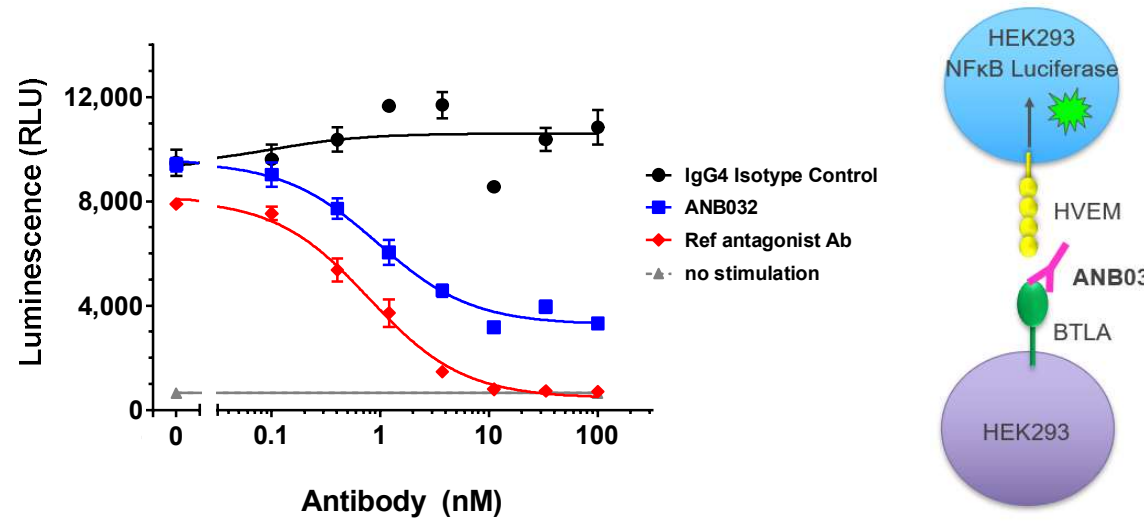
Luminescence (RLU)
12,000
8,000
4,000
0
0
0.1
1
10
100
Antibody (nM)
IgG4 Isotype Control
ANB032
Ref antagonist Ab
no stimulation
HEK293 NFκB Luciferase
HVEM
ANB032
BTLA
HEK293

ANB032 inhibits HVEM signaling induced by BTLA when BTLA and HVEM are expressed in *trans*. 293c18 cells were stably transfected with full length huHVEM and the NF-κB-luciferase reporter vector to provide a readout of HVEM signaling. HVEM-dependent NF-κB signaling was initiated in *trans* by membrane BTLA expressed on 293c18 cells. Addition of ANB032 resulted in a concentration-dependent inhibition of HVEM-dependent NF-κB signaling in response to huBTLA 293c18 cells. Maximal inhibition at the highest concentrations of ANB032 tested was ~70%.

Figure 4: ANB032 preserves *trans* HVEM-induced BTLA signaling

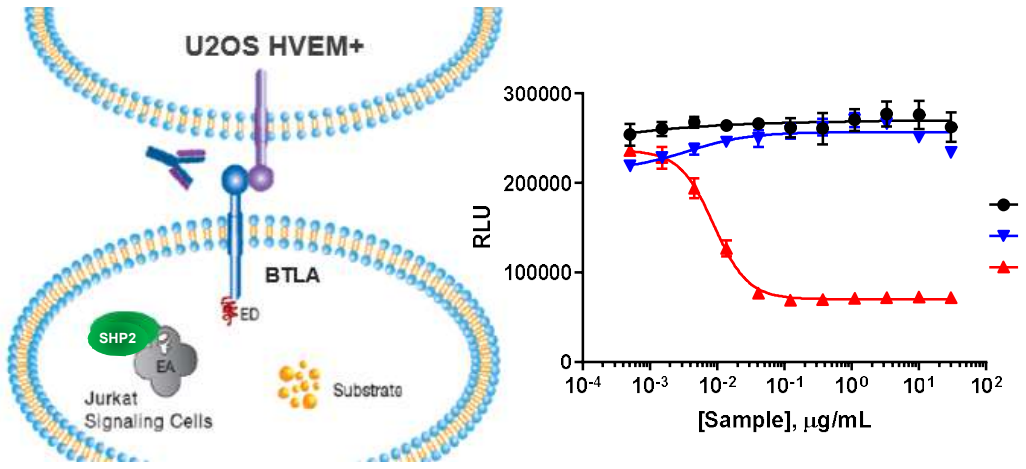
U2OS HVEM+
BTLA
ED
SHP2
EA
Jurkat Signaling Cells
Substrate
RLU
300000
200000
100000
0
10^{-4}
10^{-3}
10^{-2}
10^{-1}
10^{0}
10^{1}
10^{2}
[Sample], μg/mL
IgG4 Isotype Control
ANB032
Ref antagonist Ab

ANB032 preserves *trans* HVEM-induced BTLA signaling in a PathHunter Jurkat BTLA assay. ANB032 was characterized for its effect on BTLA signaling (SHP2 recruitment to BTLA) in response to cell-associated HVEM using the PathHunter Jurkat BTLA signaling assay (Eurofins DiscoverX). HVEM ligand was presented in *trans* on human HVEM transfected U-2 OS osteosarcoma cell line to initiate signaling. Addition of ANB032 had no effect on SHP2 recruitment to BTLA in response to cell-associated HVEM, consistent with the finding that ANB032 does not inhibit soluble HVEM-Fc binding to BTLA expressing cells and demonstrates that ANB032 does not inhibit HVEM signaling through BTLA in *trans*. These data suggests that ANB032 binds to BTLA in such a way that HVEM-mediated signaling is inhibited, but not BTLA phosphorylation mediated by HVEM. In contrast, addition of the reference BTLA antagonist antibody resulted in potent inhibition of BTLA signaling in response to cell-associated HVEM, consistent with its ability to strongly inhibit HVEM binding to BTLA.

Figure 5: ANB032 in solution is capable of directly inducing BTLA signaling, that may be potentiated by Fc receptor engagement

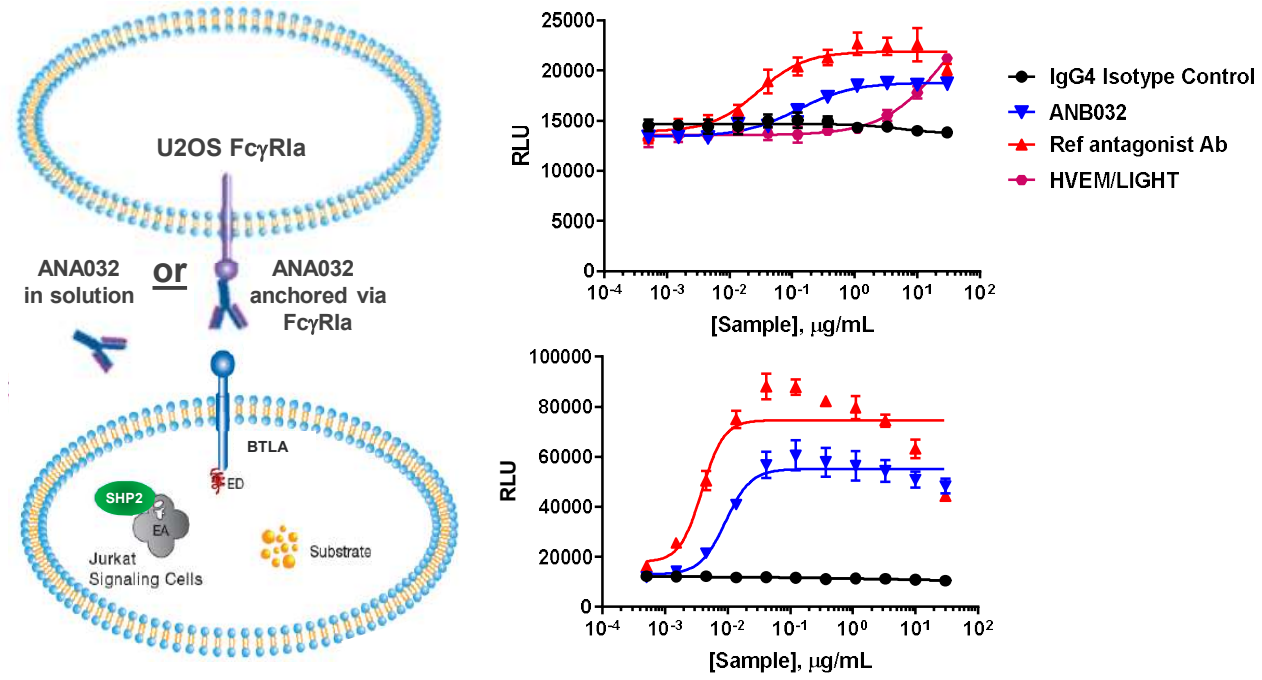
U2OS FcγRIa
ANA032 in solution
or
ANA032 anchored via FcγRIa
BTLA
ED
SHP2
EA
Jurkat Signaling Cells
Substrate
RLU
25000
20000
15000
10000
5000
0
[Sample], μg/mL
IgG4 Isotype Control
ANB032
Ref antagonist Ab
HVEM/LIGHT
100000
80000
60000
40000
20000
10^{-4}
10^{-3}
10^{-2}
10^{-1}
10^{0}
10^{1}
10^{2}

The ability of ANB032 to directly induce BTLA signaling was assessed in the PathHunter Jurkat BTLA assay. The first assay format that was used assessed the direct agonistic activity of ANB032 in solution, which resulted in induction of BTLA signaling. An IgG4 isotype control antibody had no SHP2 recruitment activity. Addition of the reference BTLA antagonist antibody also resulted in BTLA signaling. The results of this experiment demonstrate that ANB032 binding to BTLA is sufficient to directly induce SHP2 recruitment to the cytoplasmic domain of BTLA which could contribute to the inhibitory function of BTLA in activated T and B cells.

The second assay assessed the agonistic activity of ANB032 for inducing BTLA signaling in the presence of U-2OS cells stably transfected with human FcγRIa to provide FcγR engagement and cluster the antibodies tested. In this assay format, addition of ANB032 resulted in potent induction of BTLA signaling. An IgG4 isotype control antibody showed no SHP2 recruitment activity. The reference BTLA antagonist antibody also showed potent induction of BTLA signaling. The results of this experiment demonstrate that FcγRIa-bound ANB032 is capable of directly inducing SHP2 recruitment to the BTLA cytoplasmic domain more efficiently than soluble ANB032. Therefore, while soluble ANB032 may initiate inhibitory signaling in activated T and B cells, the potential for FcγRIa-bound ANB032 to induce direct inhibitory signaling may be enhanced.

Figure 6: Efficacy of ANB032 in a humanized murine GvHD model

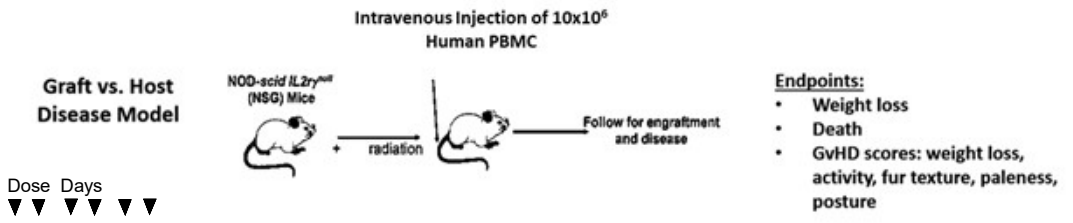
Intravenous Injection of 10x10^6 Human PBMC
Graft vs. Host Disease Model
NOD-scid IL2rγ[null] (NSG) Mice
radiation
Follow for engraftment and disease
Endpoints:
Weight loss
Death
GvHD scores: weight loss, activity, fur texture, paleness, posture

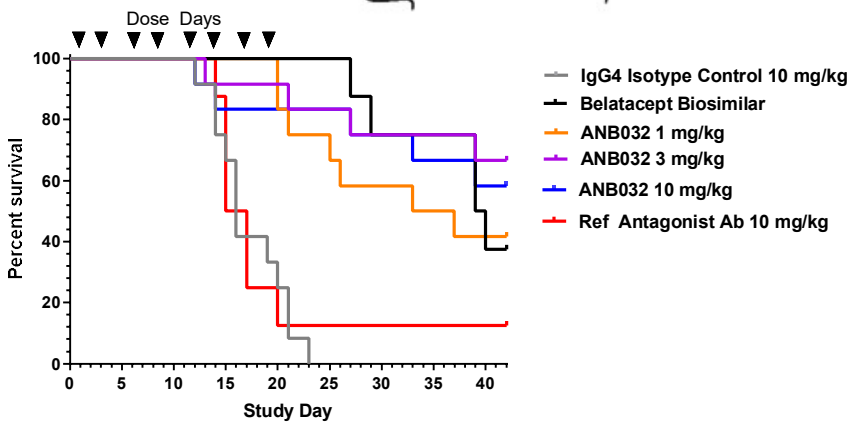
Dose Days
Percent survival
100
80
60
40
20
0
0
5
10
15
20
25
30
35
40
Study Day
IgG4 Isotype Control 10 mg/kg
Belatacept Biosimilar
ANB032 1 mg/kg
ANB032 3 mg/kg
ANB032 10 mg/kg
Ref Antagonist Ab 10 mg/kg

Treatment	Median Survival
IgG4 Isotype, 10 mg/kg	16 days
Belatacept biosimilar	39.5 days
ANB032, 1 mg/kg	35 days
ANB032, 3 mg/kg	Not defined
ANB032, 10 mg/kg	Not defined
Reference Antagonist Ab	16 days

The *in vivo* efficacy of ANB032 was evaluated in a xenogeneic humanized mouse model of acute graft versus host disease (GvHD). This model is a T cell-mediated disease induced by engraftment of human PBMCs in sub-lethally irradiated NSG mice. At the end of study on Day 42, animals treated with ANB032 at all doses showed statistically significant increased overall survival as compared with animals treated with isotype control antibody (10 mg/kg ANB032 dose group versus isotype, p=0.0019; 3 mg/kg ANB032 versus isotype, p=0.0002; 1 mg/kg ANB032 versus isotype, p=0.0001). There was not a significant difference in survival between the 10 mg/kg and 3 mg/kg ANB032 dose groups, suggesting that efficacy in the GvHD model may be maximal at doses equal to and greater than 3 mg/kg. No survival benefit was observed in animals treated with the reference antagonist antibody compared to isotype control treated animals.

Conclusions

- **BTLA modulating antibodies that down-regulate or up-regulate HVEM signaling can be discovered and optimized for therapeutic purposes.**
- **ANB032 is a humanized IgG4 anti-BTLA antibody that does not inhibit BTLA/HVEM interactions while modulating the bidirectional signaling pathways. ANB032 preserves HVEM-mediated co-inhibitory signaling through BTLA while preventing BTLA-mediated co-stimulatory signaling through HVEM. Additionally, ANB032 has the capacity to directly induce co-inhibitory agonistic signaling through BTLA, which is potentiated by Fc receptor engagement.**
- **ANB032 demonstrated efficacy in a xenogeneic NSG-Human-PBMC graft vs. host disease model, while a reference anti-BTLA antagonist antibody that disrupted BTLA/HVEM interactions demonstrated no efficacy, highlighting the critical role of BTLA/HVEM modulation by ANB032 as a key mechanism in regulating this T cell-dependent disease.**
- **Anti-BTLA antibodies that regulate BTLA/HVEM interactions to down-modulate T-cell responses have the potential to restore and maintain immune balance in autoimmune and inflammatory diseases.**