PO558

A Phase 2b, Randomized, Double-blind, Placebo-controlled, Multicenter, Global Study to Evaluate the Efficacy and Safety of ANB032 in the Treatment of Subjects with Moderate to Severe Atopic Dermatitis

Jonathan Silverberg[1], Benjamin Ehst[2], Bart Burington[3*], Paul Lizzul[3], Kenneth Luu[3], Jocelyne Papacharalambous[3], Priya Raina[3], Bruce Randazzo[3], Cailin Sibley[3], Emma Guttman-Yassky[4]
[1]The George Washington University School of Medicine and Health Sciences, Washington DC, USA; [2]Oregon Medical Research Center, Portland, OR, USA; [3]AnaptysBio, Inc., San Diego, CA, USA; [4]Icahn School of Medicine at Mount Sinai, New York, NY, USA

INTRODUCTION

Unmet Need in Atopic Dermatitis (AD)

- AD is a systemic and heterogenous disease, and its pathophysiology is not well understood
- Although there has been an increase in effective treatment options, patient satisfaction with treatment is only partially or not met in more than 75% of patients surveyed with moderate to severe AD[1]
- Current treatment options do not target all the diverse immune cells and cytokines that contribute to the pathogenesis of this disease

B and T Lymphocyte Attenuator (BTLA)

- BTLA is a co-inhibitory checkpoint receptor that is expressed across a range of immune cells, including T cells (Th1, Th2, Th17, Th22), B cells, and dendritic cells (DCs), which are key cell types contributing to inflammatory diseases such as AD
 - T cell activation requires antigen presentation in the context of MHC to the TCR, and co-stimulation via CD80/86 to CD28
- BTLA engagement inhibits T cell proliferation and reduces secretion of inflammatory cytokines
- BTLA engagement on DCs modulates maturation and function, by reducing both antigen presentation by MHC and expression of costimulatory molecules (**Figure 1**)

Figure 1. BTLA is an inhibitory checkpoint receptor

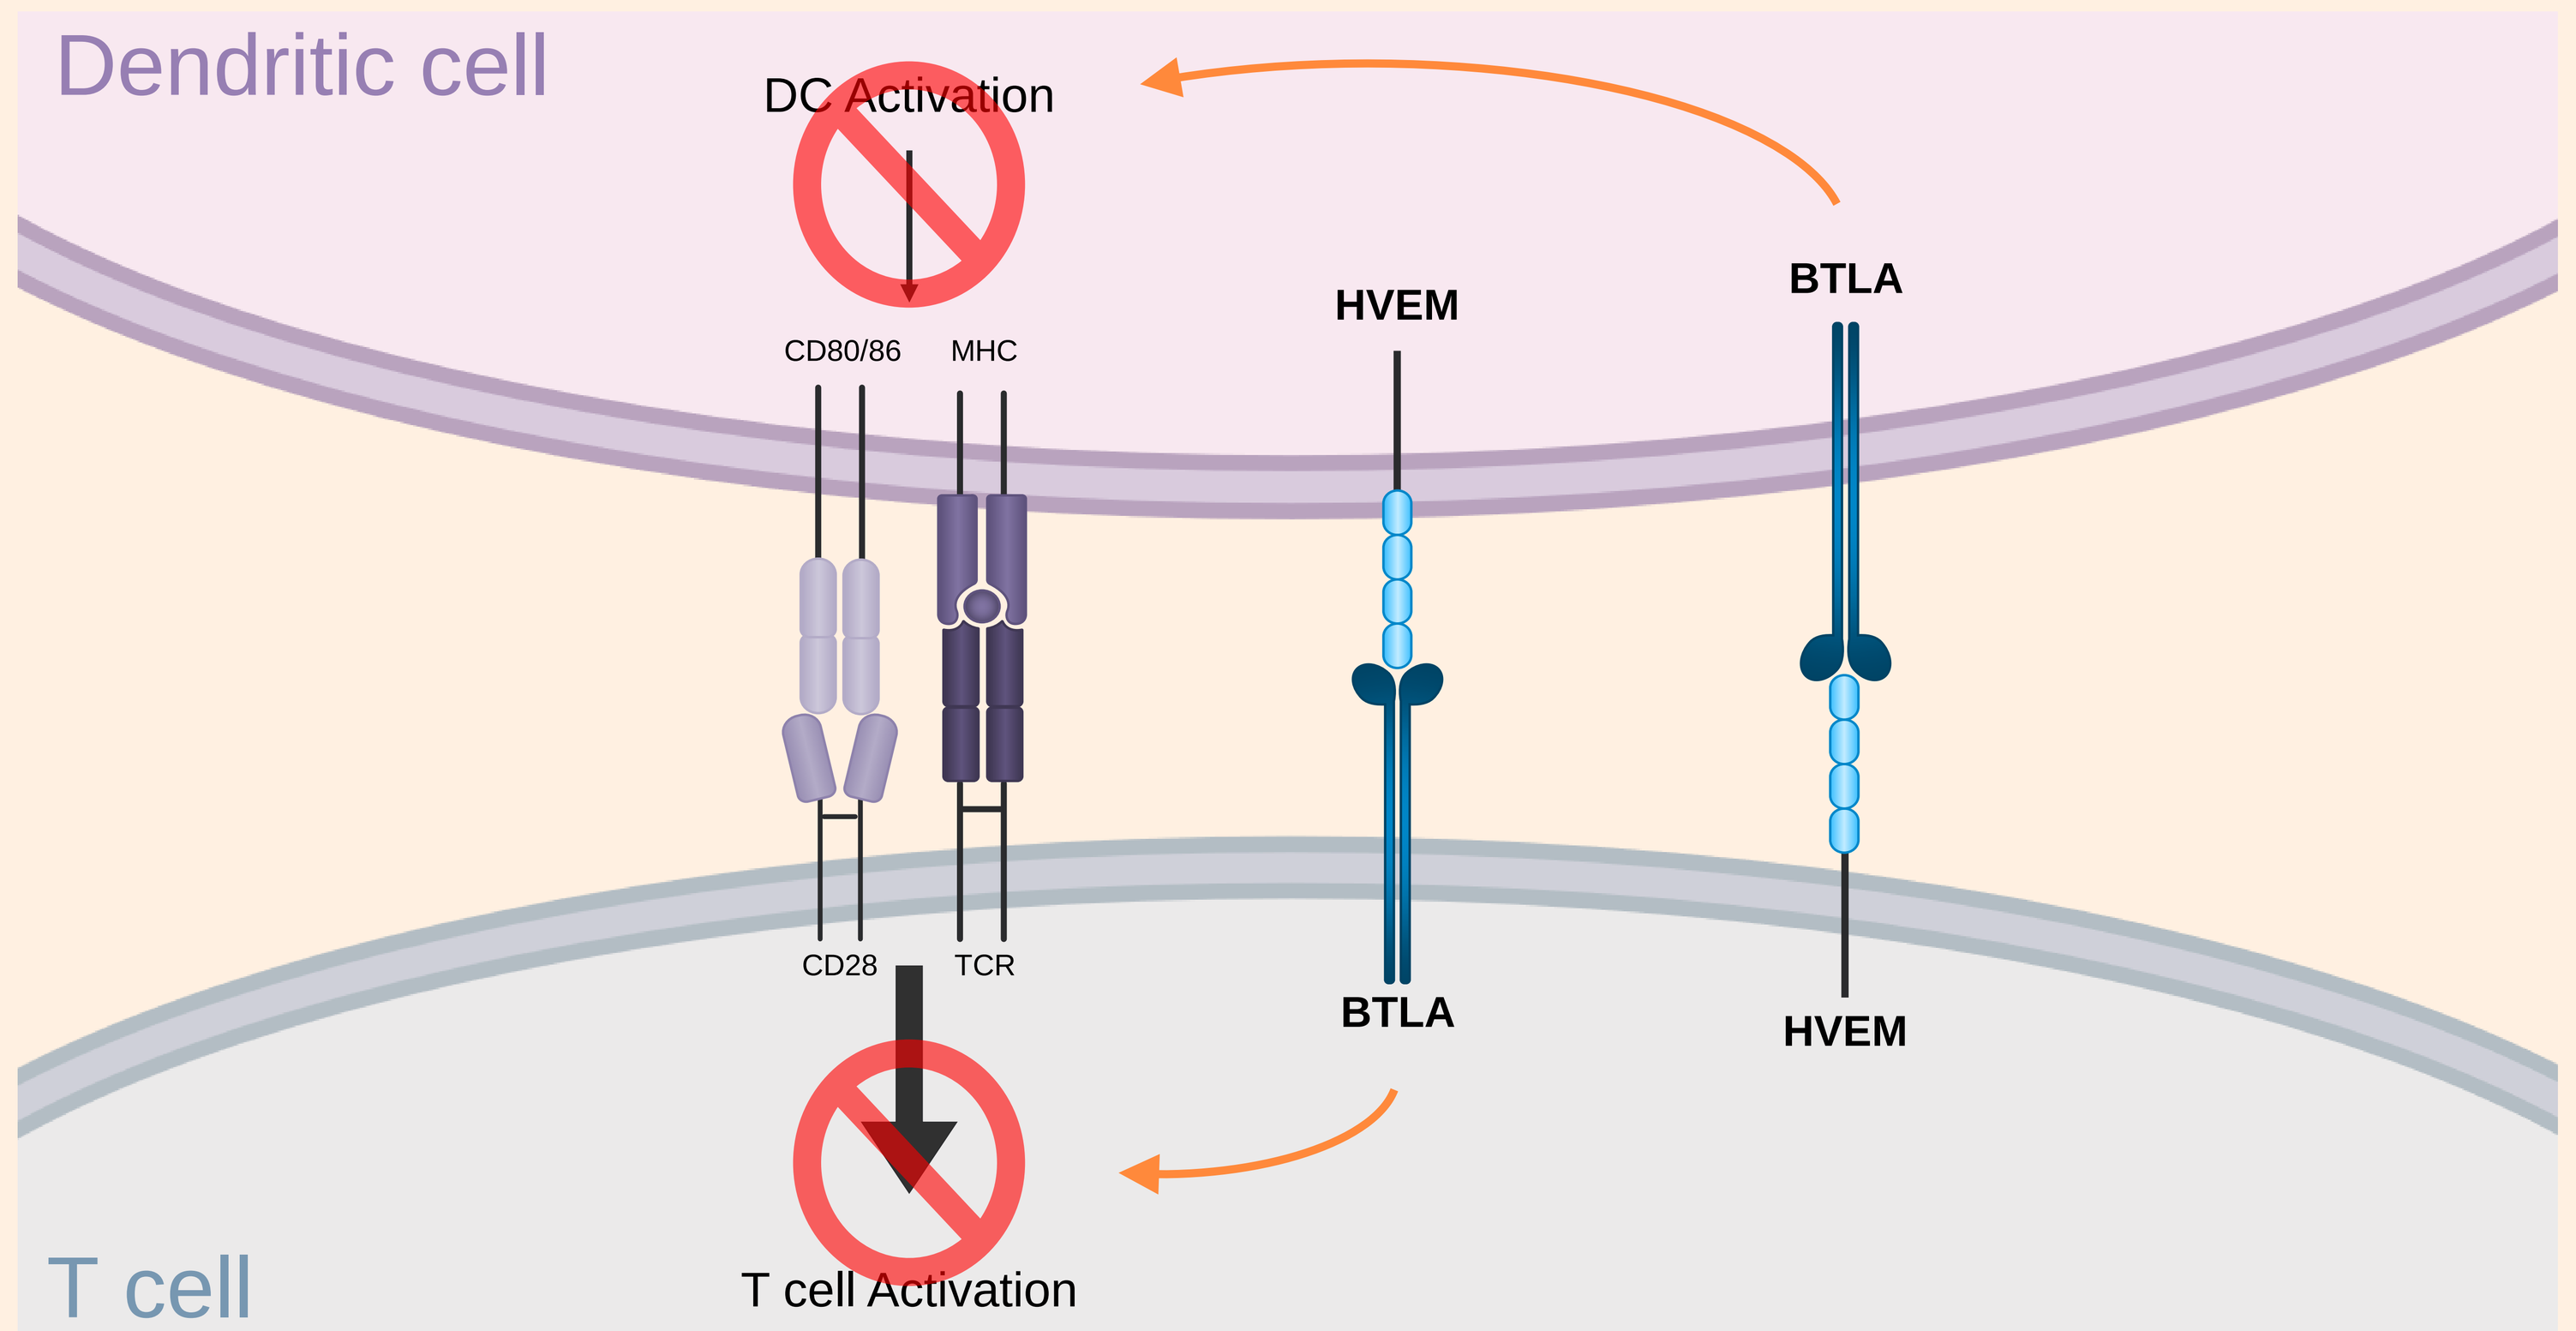
Dendritic cell
DC Activation
CD80/86
MHC
HVEM
BTLA
CD28
TCR
BTLA
HVEM
T cell Activation
T cell

ANB032, a BTLA Agonist Antibody

- ANB032 is a humanized IgG4/κ monoclonal antibody to human BTLA that modulates BTLA activity in a non-competitive fashion with its ligand, HVEM
- ANB032 binds to BTLA on Th1, Th2, Th17, and Th22 T cells and DCs and augments BTLA agonism (**Figure 2**)
- ANB032's agonist signal modulates DC maturation and function, reducing antigen presentation and expression of co-stimulatory molecules, including inducing Tregs[2]
- Cytokine secretion (Th1, Th2, Th17, & Th22) was inhibited in AD patient-derived PBMCs exposed to ANB032[3]
- In a Phase 1 first-in-human healthy volunteer study, ANB032 was well-tolerated, had a favorable PK profile, demonstrated robust target engagement and a partial reduction in BTLA expression, consistent with observations in animal models of dermatitis and inflammation (Luu, et al. *EADV 2023 Abstract 5603, being presented on Oct. 13 at 8:30 am CET*)
- The robust preclinical data and Phase 1 trial results support advancing the clinical development of ANB032 in a Phase 2b study in AD (ARISE AD; NCT05935085)

OBJECTIVE

ARISE-AD is evaluating the safety, tolerability, and efficacy of ANB032 in subjects with moderate to severe AD

METHODS

Study Design and Subjects

- This global Phase 2b study is a randomized, double-blind, placebo-controlled, parallel-group, multicenter trial (**Figure 3**)
 - Male and female age 18 to 65 years with moderate to severe AD for at least 6 months with a history of inadequate response to topical AD medications or unable to tolerate topical treatment
 - At screening and randomization: EASI ≥ 16; vIGA-AD ≥ 3; AD-involved BSA ≥ 10%
 - Subjects who are either dupilumab/IL-13 blocker-naïve or -experienced are eligible for enrolment

Treatment

- Approximately 160 subjects are being randomized in a 1:1:1:1 ratio in 4 equal treatment groups, evaluating 3 subcutaneous (SC) doses and schedules of ANB032 and placebo
- The treatment period is 14 weeks with an additional 12-weeks of follow up

Figure 2. ANB032 Proposed Mechanism of Action

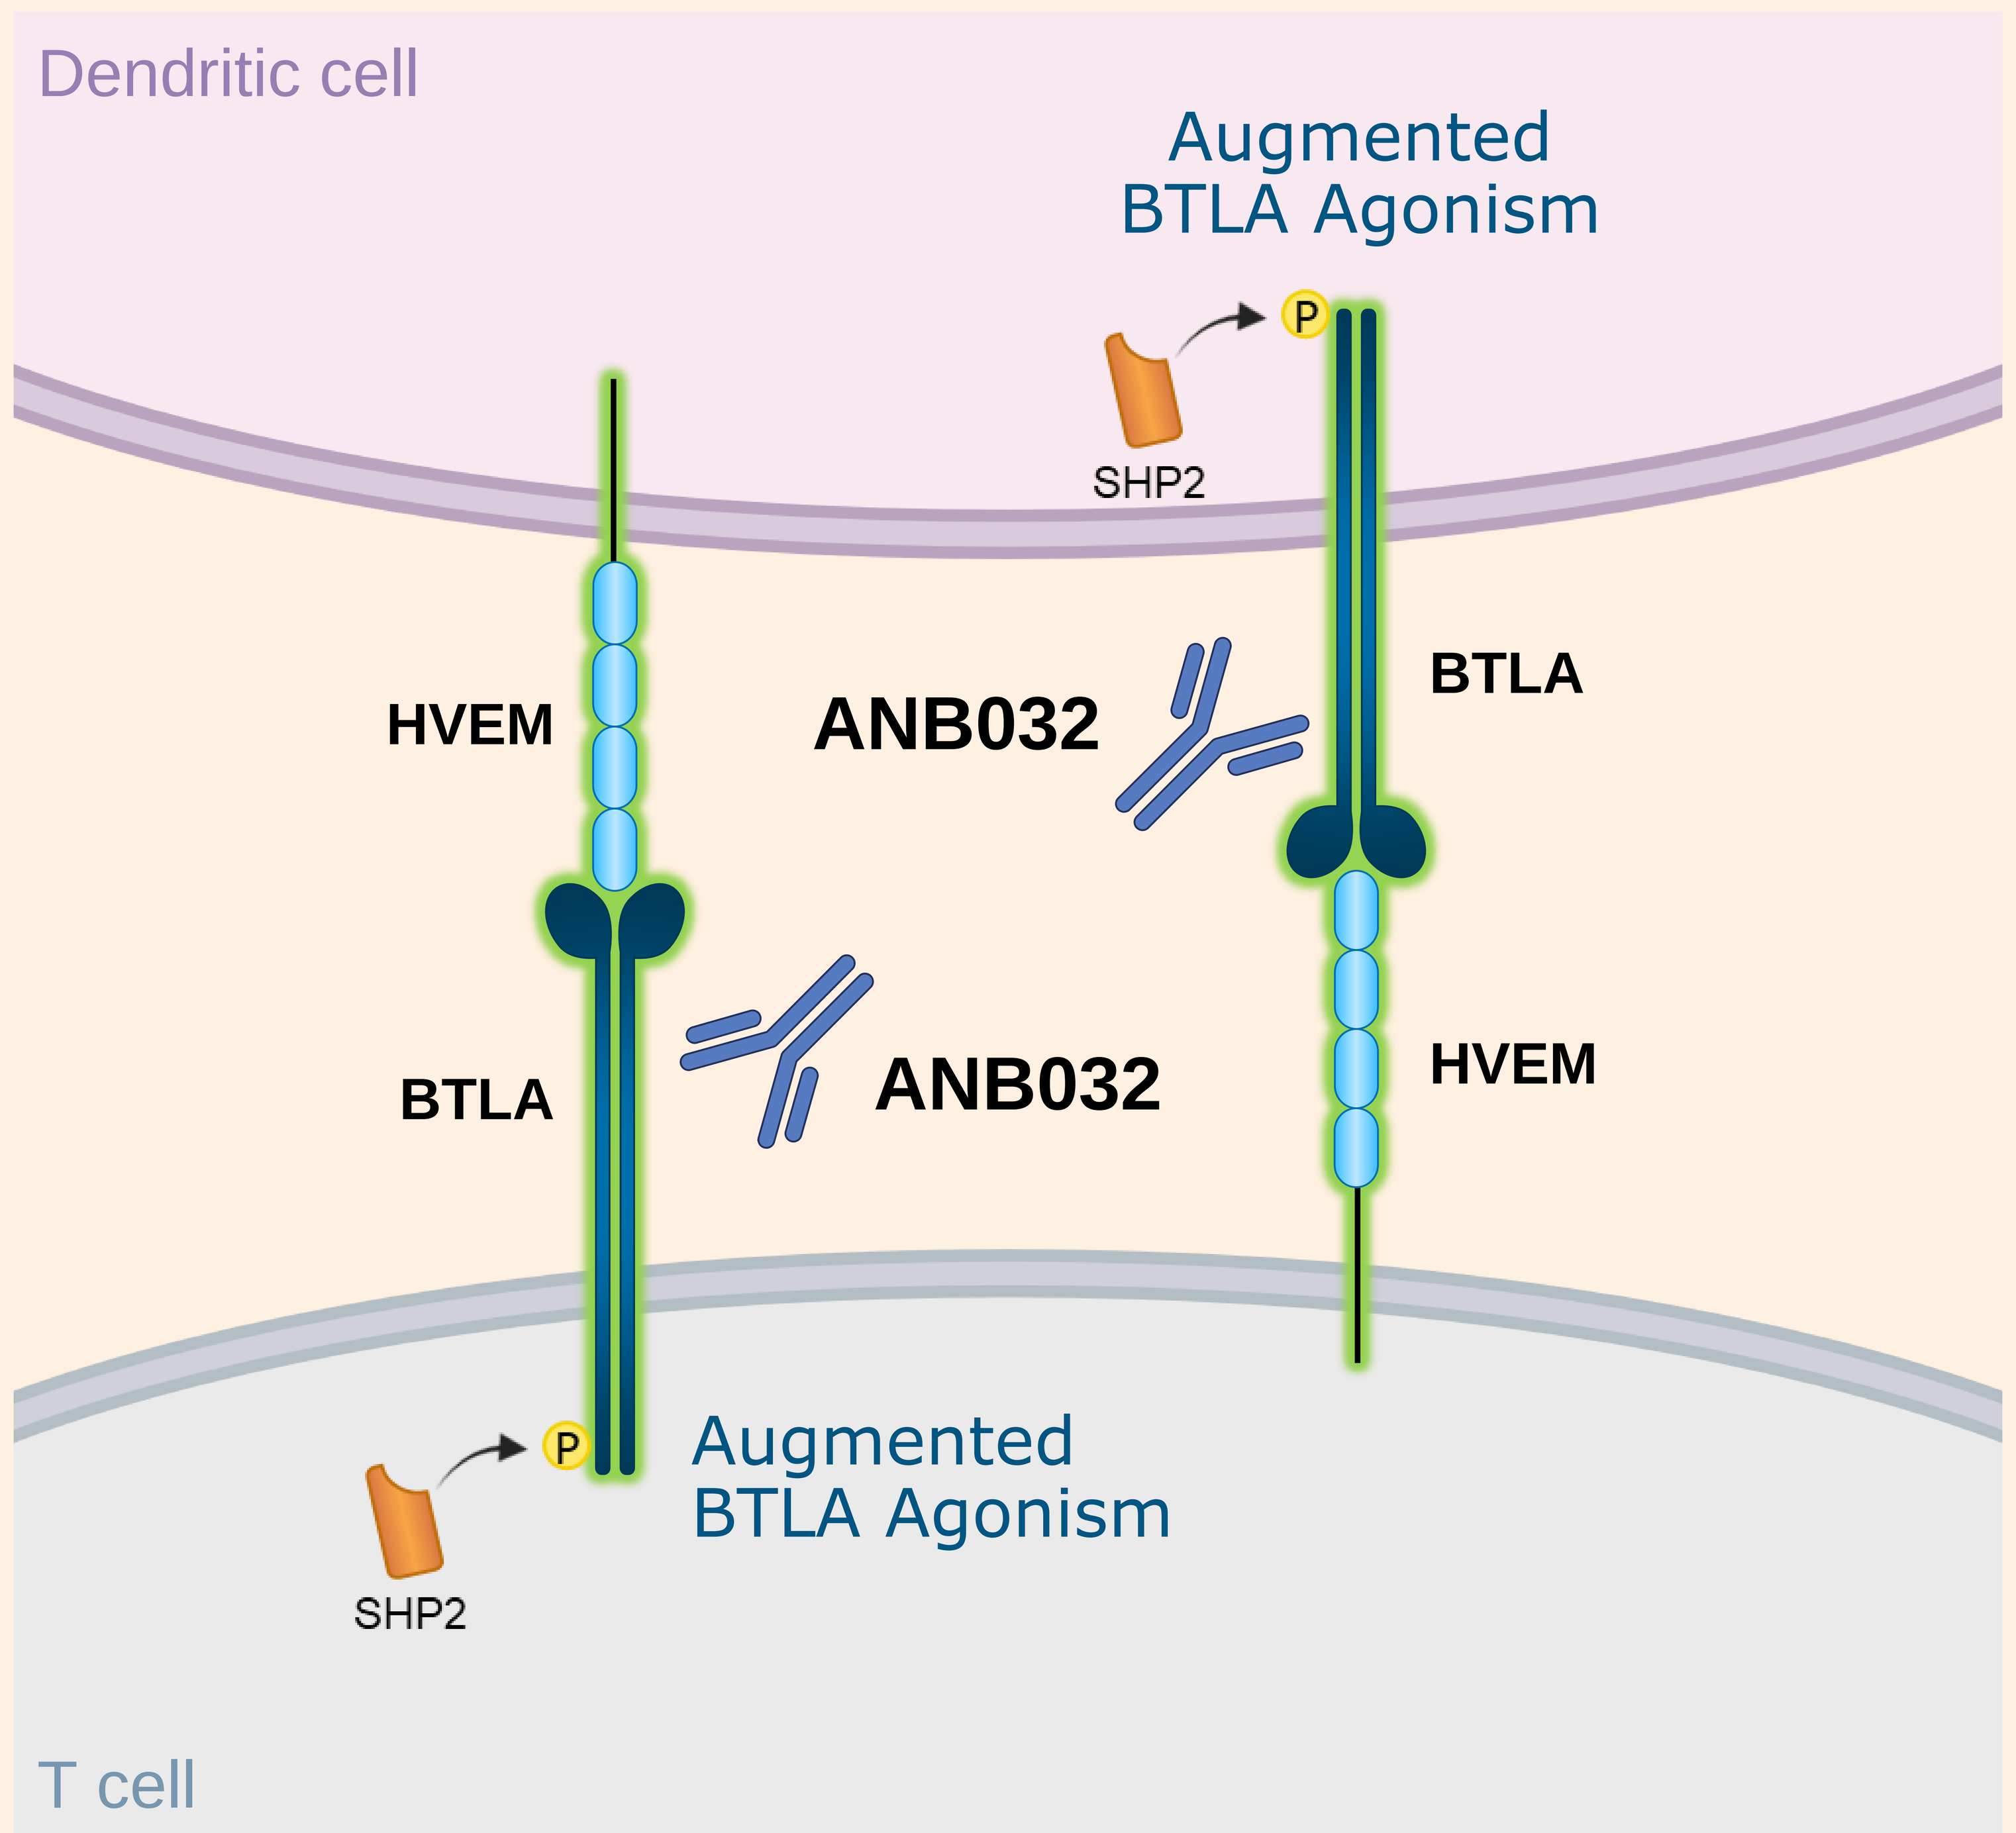
Dendritic cell
Augmented BTLA Agonism
P
SHP2
HVEM
ANB032
BTLA
BTLA
ANB032
HVEM
P
SHP2
Augmented BTLA Agonism
T cell

Figure 3. ARISE-AD Study Design

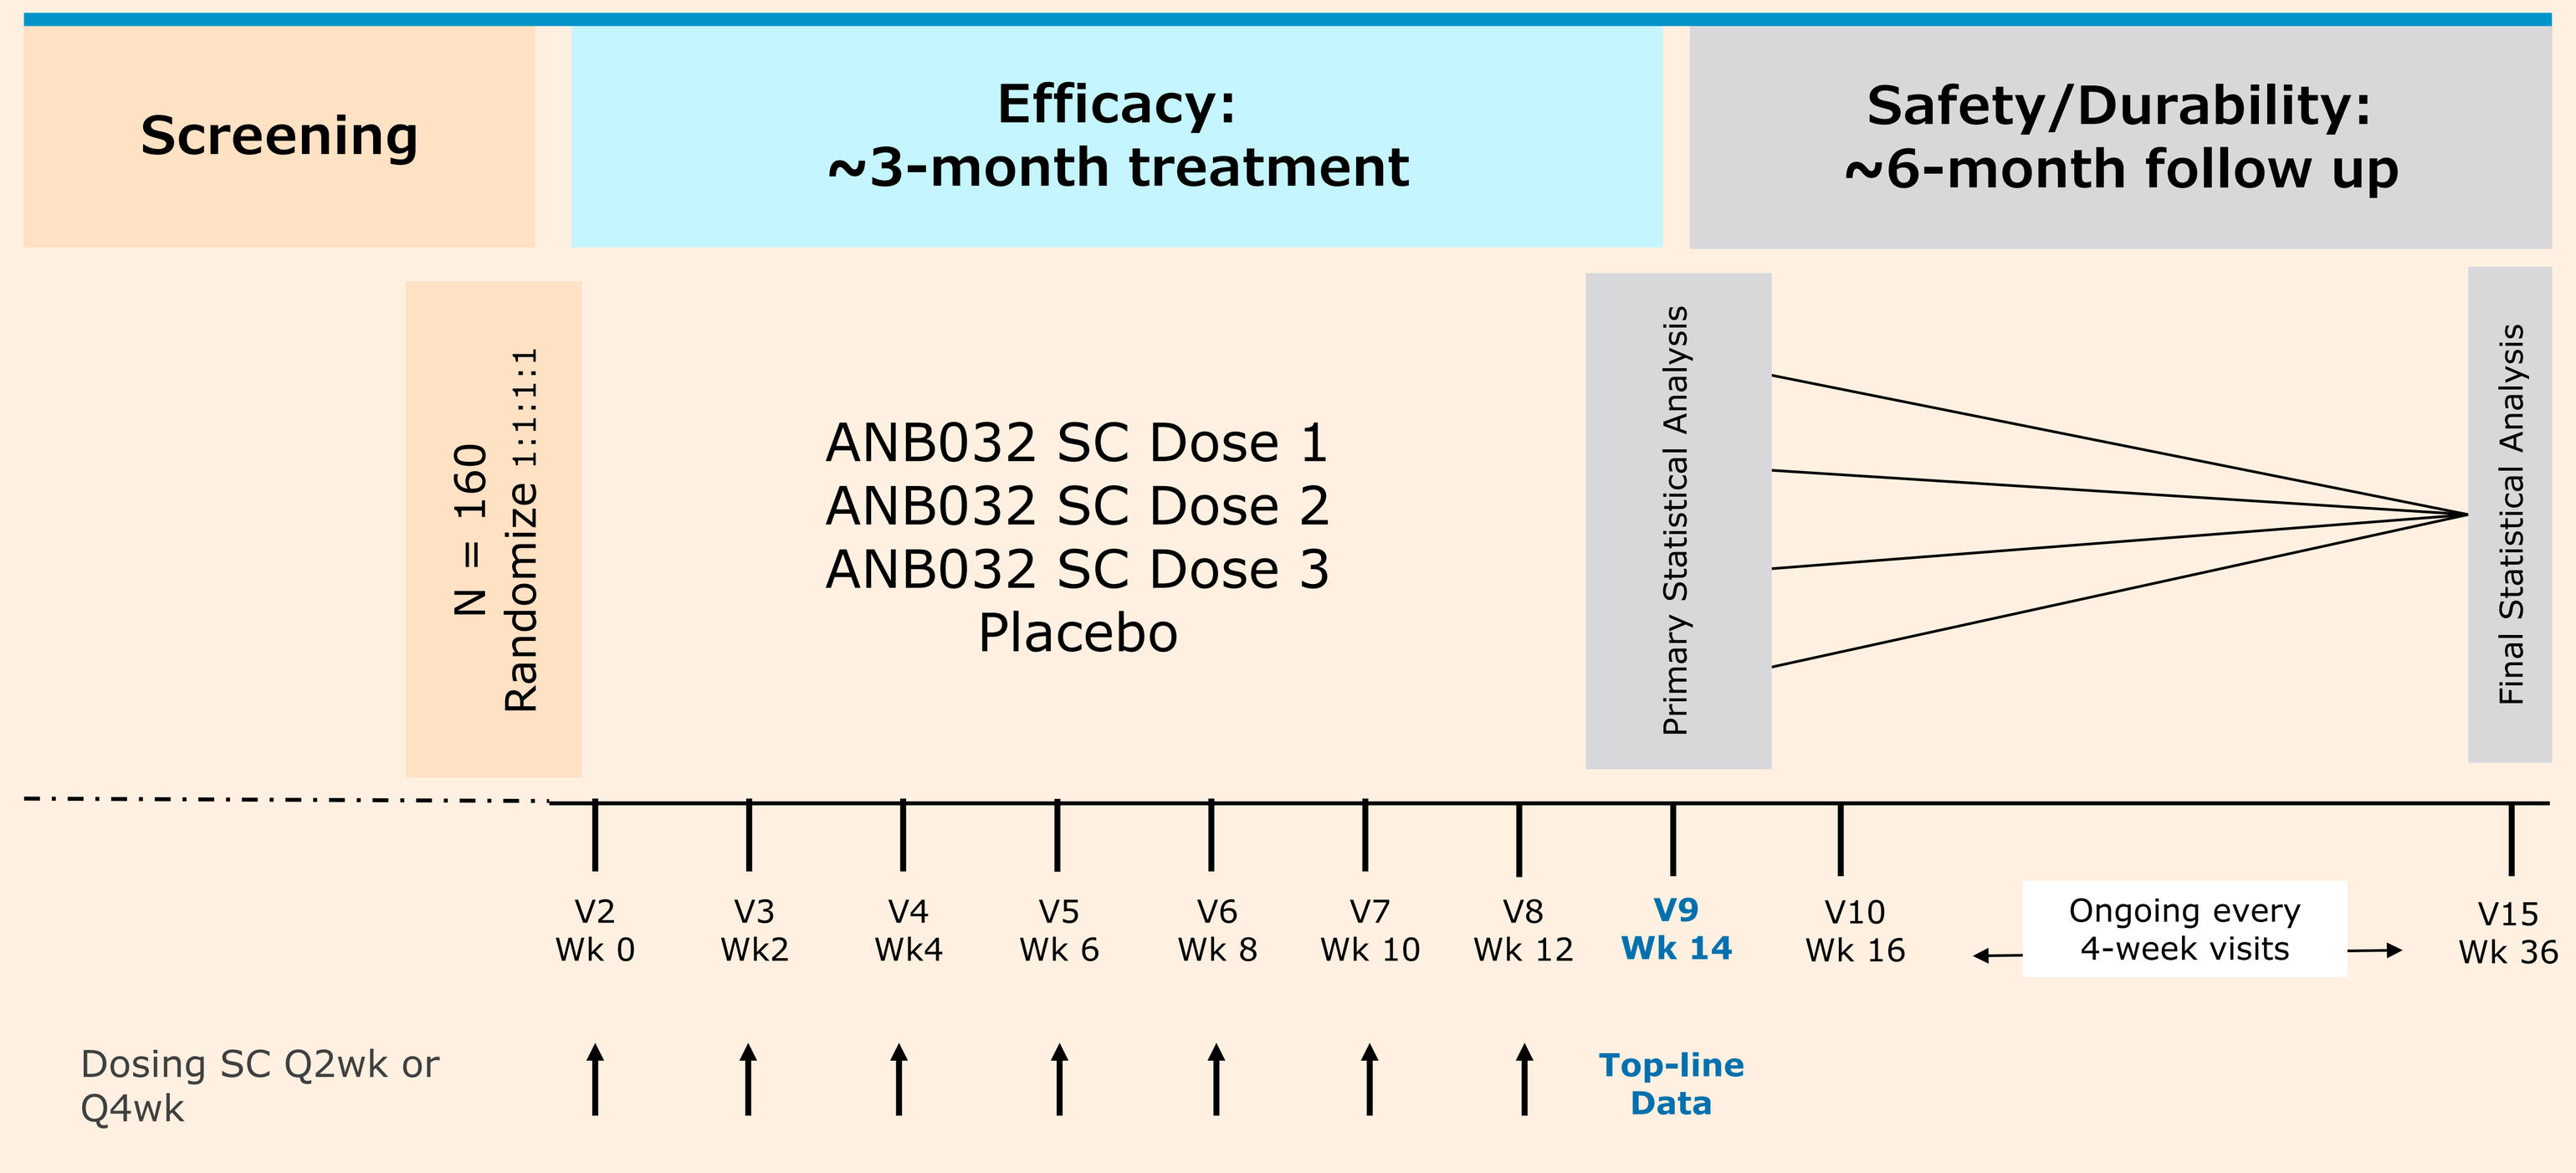
Screening
Efficacy: ~3-month treatment
Safety/Durability: ~6-month follow up
N = 160
Randomize 1:1:1:1
ANB032 SC Dose 1
ANB032 SC Dose 2
ANB032 SC Dose 3
Placebo
Primary Statistical Analysis
Final Statistical Analysis
V2 Wk 0
V3 Wk2
V4 Wk4
V5 Wk 6
V6 Wk 8
V7 Wk 10
V8 Wk 12
V9 Wk 14
V10 Wk 16
Ongoing every 4-week visits
V15 Wk 36
Dosing SC Q2wk or Q4wk
Top-line Data

Patient population		• Adults with moderate-to-severe atopic dermatitis • Include both dupilumab/IL-13 naïve and experienced patients
Endpoints	Primary	• Mean change from Baseline in EASI at Week 14
	Secondary	• EASI-75 • vIGA-AD 0 (clear) or 1 (almost clear) and a ≥ 2-point reduction (improvement) • PNRS (itch), DLQI, SCORAD • Safety
Exploratory endpoints		• Th1/Th2/Th17 cytokines and other soluble biomarkers, tissue samples (tape strips, biopsies)

DISCUSSION

- Current treatment options in AD do not target all the diverse immune cells and cytokines that contribute to the pathogenesis of this disease
- BTLA is a coinhibitory checkpoint receptor that modulates activation of T cells, B cells, and DCs
- The synergistic effect of ANB032's direct agonism of T cells and DCs suggests its potential to modulate all phases of the pathogenic inflammatory response in diseases such as AD
- ARISE-AD, a Phase 2b study of ANB032, will add to the evolving treatment landscape of AD and further the understanding of the pathophysiology of this heterogenous disease
- This study was initiated in Q2 2023. Topline data are expected by year end 2024

ACKNOWLEDGEMENTS

- This research is sponsored by AnaptysBio, Inc.
- Author disclosures: JS, consultant and advisor to AnaptysBio; BE, consultant, speaker, and clinical trial investigator for AnaptysBio; EGY, consultant and received research grants from AnaptysBio; PL, KL, JP, PR, BR, CS are employees of AnaptysBio, Inc. For JS, BE, and EGY, additional disclosures available on request.

*Bart Burington is no longer employed by AnaptysBio, Inc.

REFERENCES

1. Augustin M, et al. Acta Derm Venereol 2022;102: adv00830.
2. Data on file. AnaptysBio, Inc.; abstract submitted
3. Luu K, et al. Presented at ISID 2023.

BSA, body surface area; **DLQI**, Dermatology Life Quality Index; **EASI**, Eczema Area and Severity Index; **HVEM**, Herpes Virus Entry Mediator; **MHC**, major histocompatibility complex; **PNRS**, Pruritus Numerical Rating Scale; **SC**, subcutaneous; **SCORAD**, Severity Scoring of Atopic Dermatitis; **TCR**, T cell receptor; **vIGA-AD**, Validated Investigator Global Assessment Scale for Atopic Dermatitis

Presented at European Academy of Dermatology and Venereology, Berlin, Germany, 11-14 October 2023